Delivery, Potency and Tolerability of Lipid Nanoparticles in PXB-mice

Hilda H. Au, Kyle B. Stephenson, and Sean C. Semple

Acuitas Therapeutics, Vancouver, BC, Canada

Abstract

Lipid nanoparticles (LNP) enable effective delivery of RNA-based therapeutics and have been clinically validated in approved intravenous products, including ONPATTRO®. Upon entry into the systemic circulation, LNP bind circulating apolipoprotein E (apoE) and interact with low-density lipoprotein receptors to facilitate their uptake into hepatocytes. Given their efficacy, low immunogenicity relative to other delivery vectors, and versatility in encapsulating a wide range of therapeutic modalities, there has been extensive adoption of LNP technology in the development of nucleic acid-based therapeutics, with increasing application in the areas of gene editing and gene modulation. A major challenge in evaluating the efficacy and tolerability of LNP-based therapeutics is the availability of suitable preclinical models. Notable limitations of traditional mouse models include species-specific differences in physiology, drug metabolism and excretion, and limited utility for therapeutics with human-specific targets or diseases with restricted tropism. More human-relevant preclinical models have emerged, which may help bridge the translational gap. Humanized liver chimeric mice, such as PXB-mice, have livers that are repopulated with functional human hepatocytes. Their livers express key transporters and metabolic enzymes, and unlike conventional mouse models, they have human-like lipoprotein profiles. These, along with other features, better represent key aspects of human liver physiology and function. Here, we evaluated the delivery, distribution, potency and tolerability of three different LNP in PXB-mice to characterize these mice as a model for LNP-mediated delivery and activity. Mice were injected intravenously with different LNP (identified as LNP07, LNP09 and LNP13) encapsulating IgG mRNA, and their relative potency was assessed by serum IgG expression. LNP13 exhibited the greatest activity of the three LNP and an expanded dose titration was performed, with doses up to 5.0 mg/kg being tolerated with no observed clinical signs. Interestingly, immunohistochemistry staining of liver sections from LNP-treated mice revealed earlier IgG protein translation in mouse hepatocytes; however, this differential in expression relative to human hepatocytes was reduced at later time points. To further investigate this, we performed pharmacokinetic analyses and MALDI mass spectrometry imaging (MSI) to visualize and quantify the distribution of LNP-derived ionizable lipid in LNP-treated mouse livers. MSI revealed extensive LNP distribution throughout the liver across both mouse and human compartments. This was corroborated by RNA in situ hybridization, which similarly showed widespread mRNA distribution at early time points post-dose. Altogether, these data suggest a potential delay in the uptake and/or expression of mRNA-LNP in human hepatocytes. Despite this, PXB-mice (and other liver-humanized mice) represent a promising preclinical model for mRNA-LNP testing and may be particularly useful in characterizing the biology of human-specific payloads when using highly active LNP.

Figure 1. mRNA-LNP with varying potencies are tolerated in PXB-mice

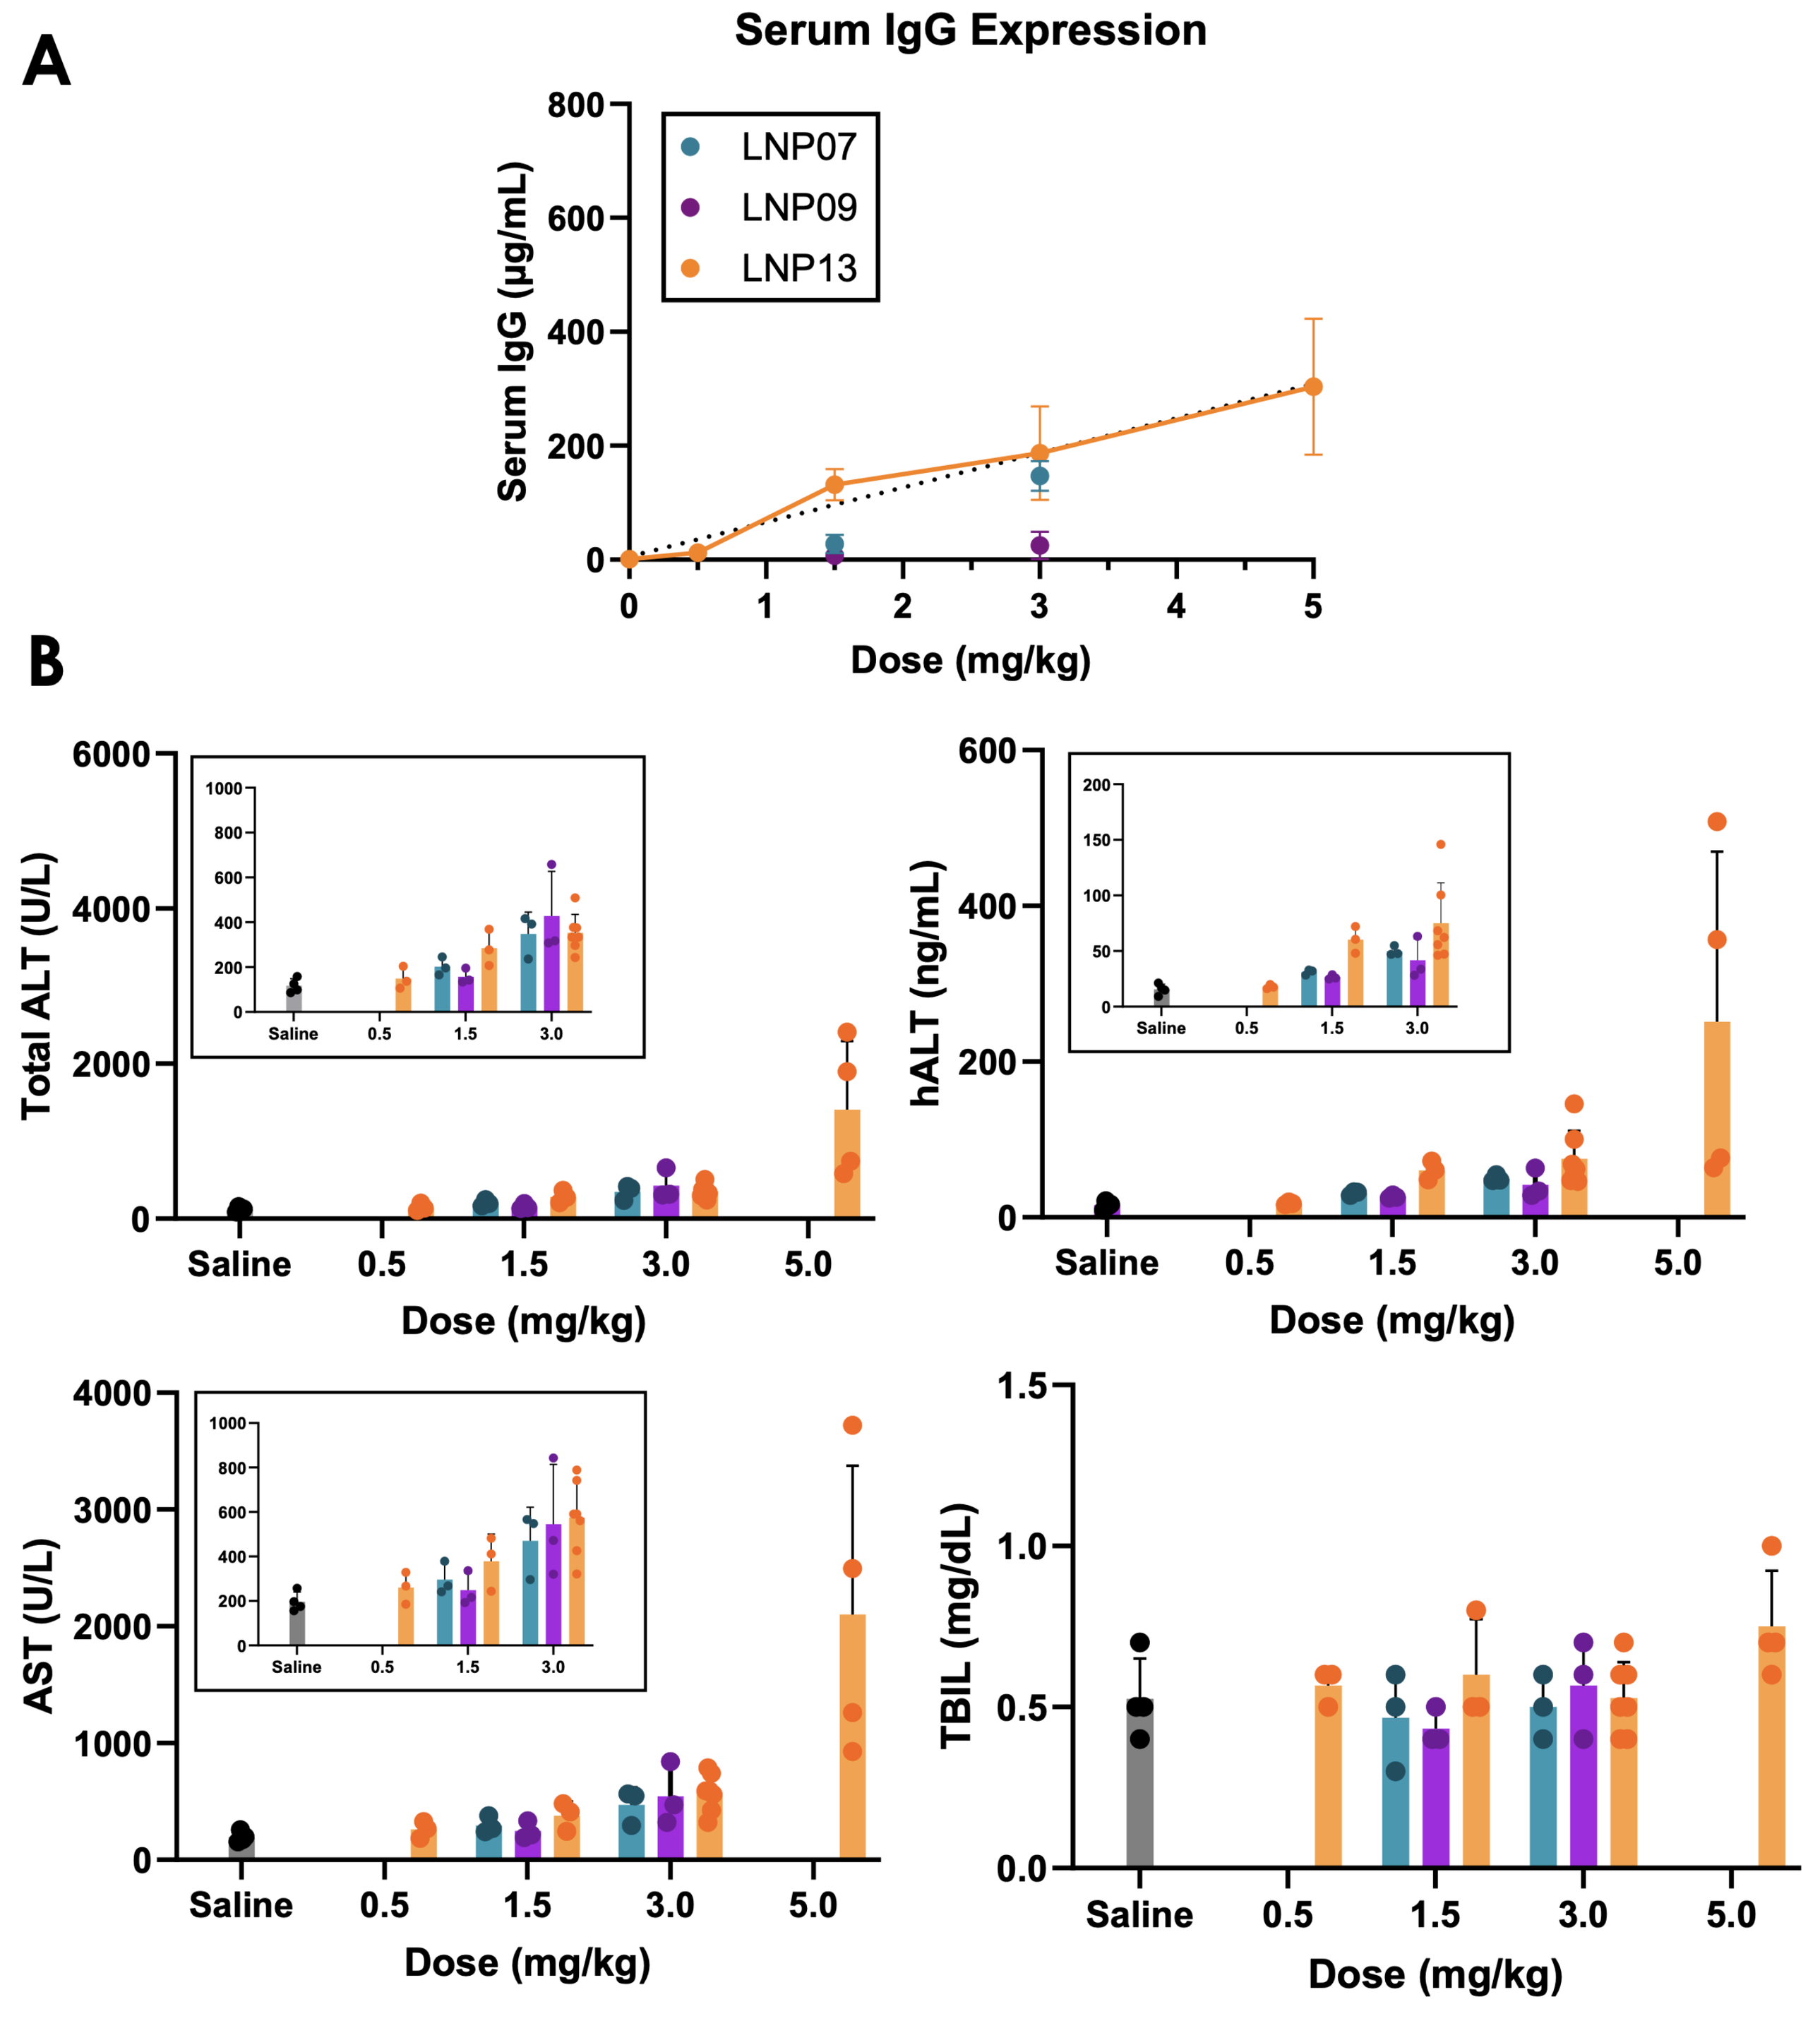
A
Serum IgG Expression
LNP07
LNP09
LNP13
Serum IgG (µg/mL)
Dose (mg/kg)
B
Total ALT (U/L)
hALT (ng/mL)
AST (U/L)
TBIL (mg/dL)
Saline
0.5
1.5
3.0
5.0
Dose (mg/kg)

Evaluating the potency of mRNA-LNP in PXB-mice. Mice were injected intravenously with 3 different LNP (LNP07, LNP09 and LNP13) encapsulating reporter IgG mRNA at different dose levels. At 24 h post-administration, blood was collected and processed to serum for IgG protein quantification (A) and clinical chemistry assessment (B). LNP potency was dose-related, with a relative rank order of LNP13>LNP07>LNP09. Dose-dependent increases in total alanine transaminase (ALT), human-specific ALT (hALT) and aspartate aminotransferase (AST) were observed. Minimal changes in total bilirubin (TBIL) levels were observed across all dose levels. LNP13 was tolerated with no observed clinical signs up to 5.0 mg/kg.

Figure 2. IgG expression was observed in human and murine hepatocytes

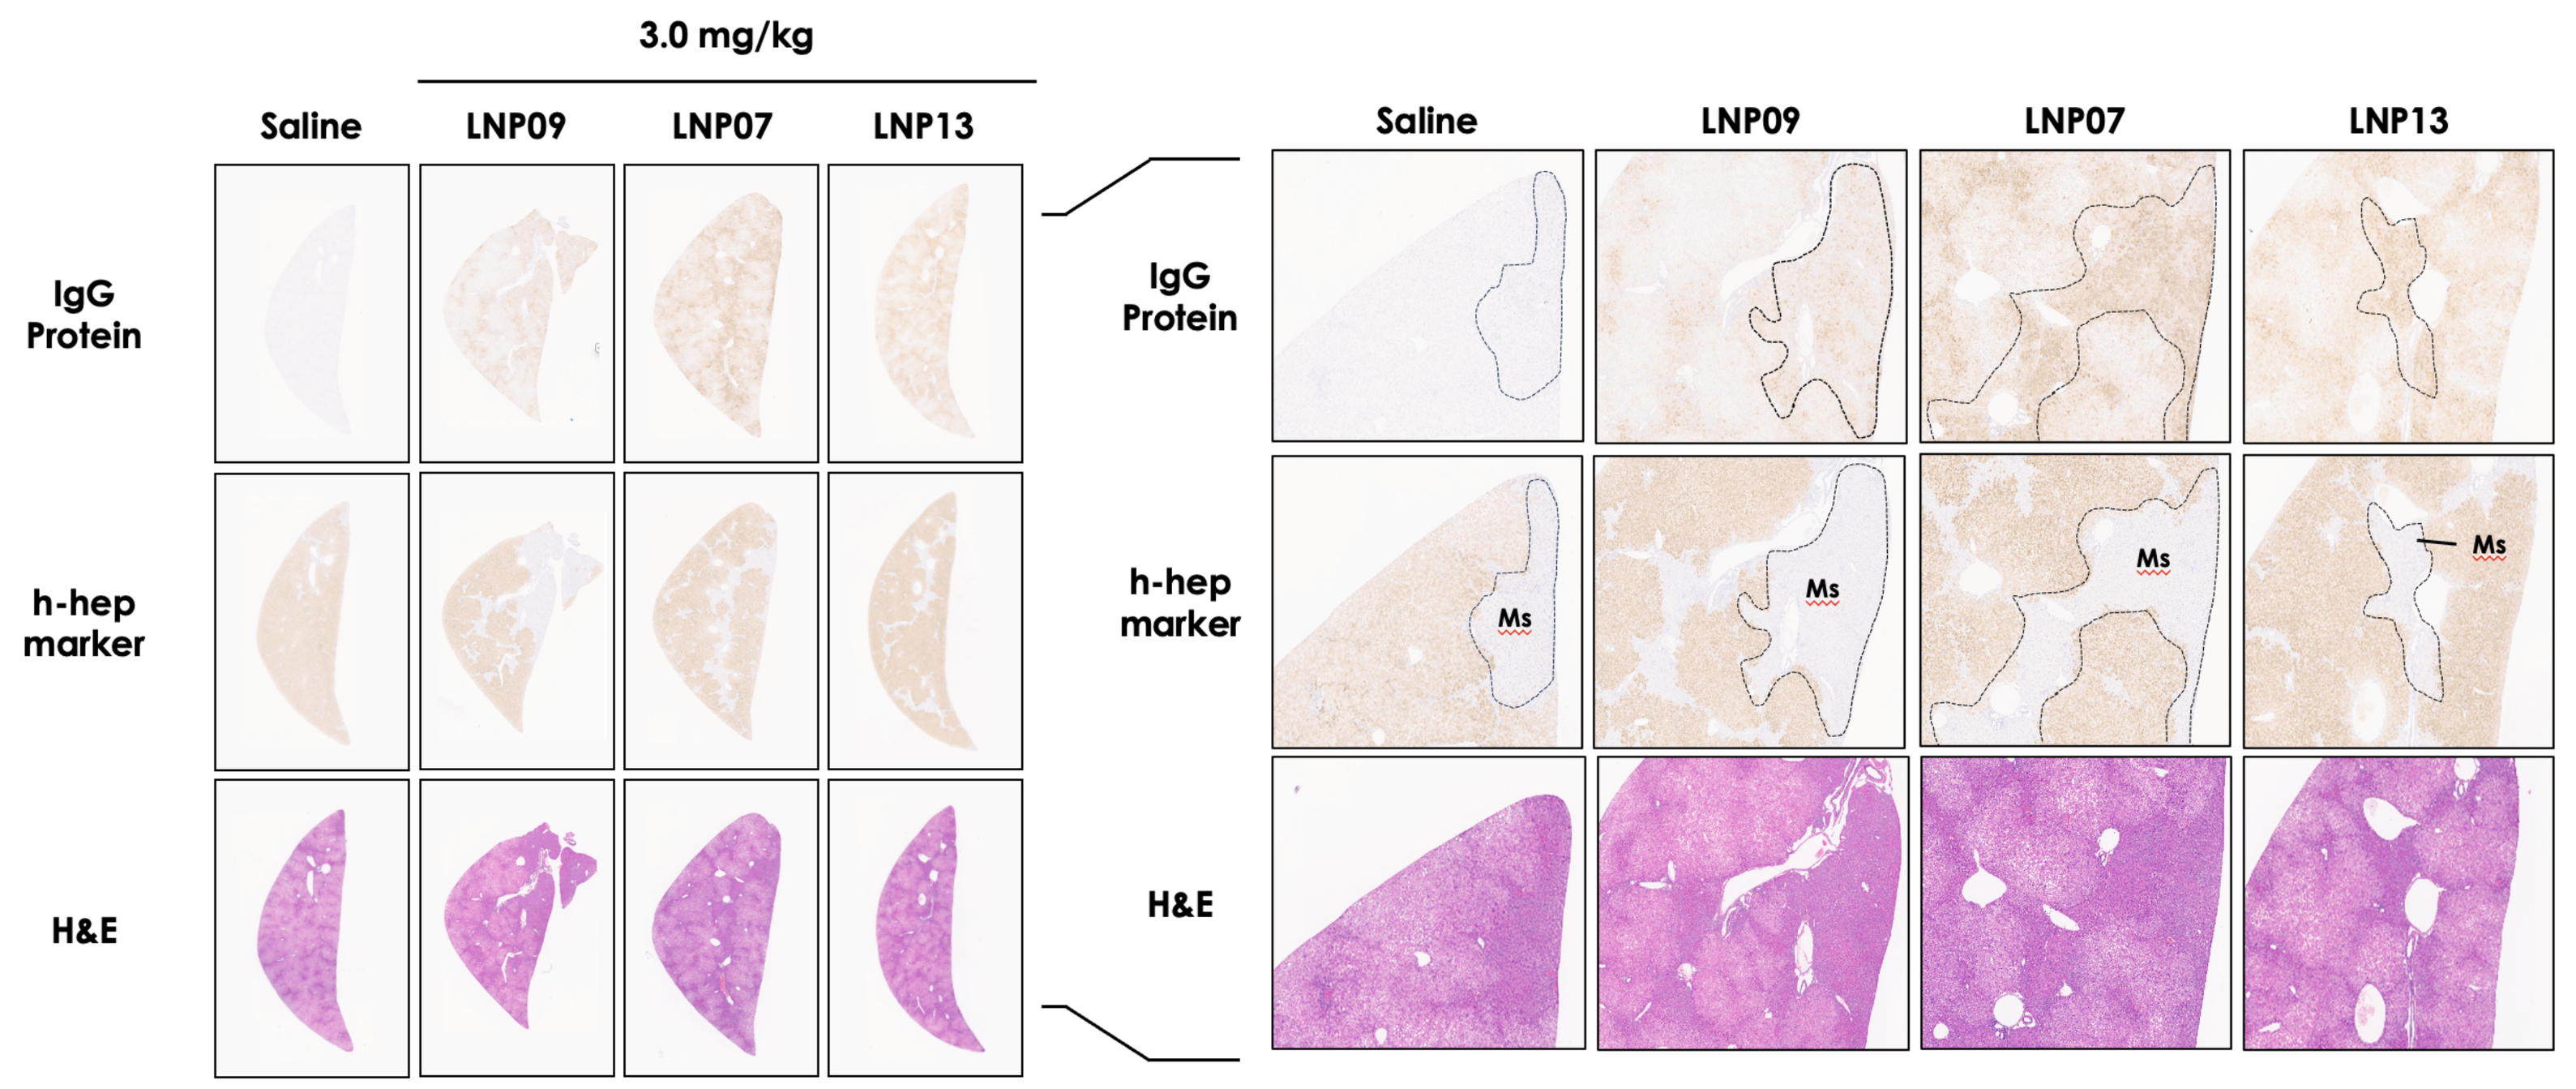
3.0 mg/kg
Saline
LNP09
LNP07
LNP13
IgG Protein
h-hep marker
H&E
Ms

Immunohistochemistry and hematoxylin-eosin (H&E) staining of serial liver sections. Liver samples were collected at 24 h post-administration of LNP09, LNP07 and LNP13 at 3.0 mg/kg and processed for staining. Human hepatocytes were identified using a human-specific marker and exhibited good IgG protein staining for LNP07 and LNP13, but diminished staining for LNP09. Overall, IgG protein expression appeared more robust in murine hepatocytes. Ms = Mouse hepatocytes

Figure 3. LNP-derived aminolipid distributes extensively through liver

Aminolipid distributes extensively across mouse liver following LNP administration. Liver sections were collected from LNP13-treated PXB-mice at 24 h post-dose and subjected to quantitative mass spectrometry imaging (QMSI)[1]. Murine (m-hep) and human hepatocytes (h-hep) were identified and segmented by immunohistochemistry (IHC) using a human-specific marker. Quantification of LNP-derived aminolipid was performed using an isotope-labelled internal standard by Multimaging™ software (Aliri Bioanalysis). Aminolipid was quantified in the entire tissue section (values in bold) and in murine hepatocytes (values in grey) and the differential was calculated to assess aminolipid concentration in human hepatocytes (values in orange). Despite observing minor differences in aminolipid concentration between murine and human hepatocytes, there is no statistically significant trend to suggest preferential accumulation in either species.

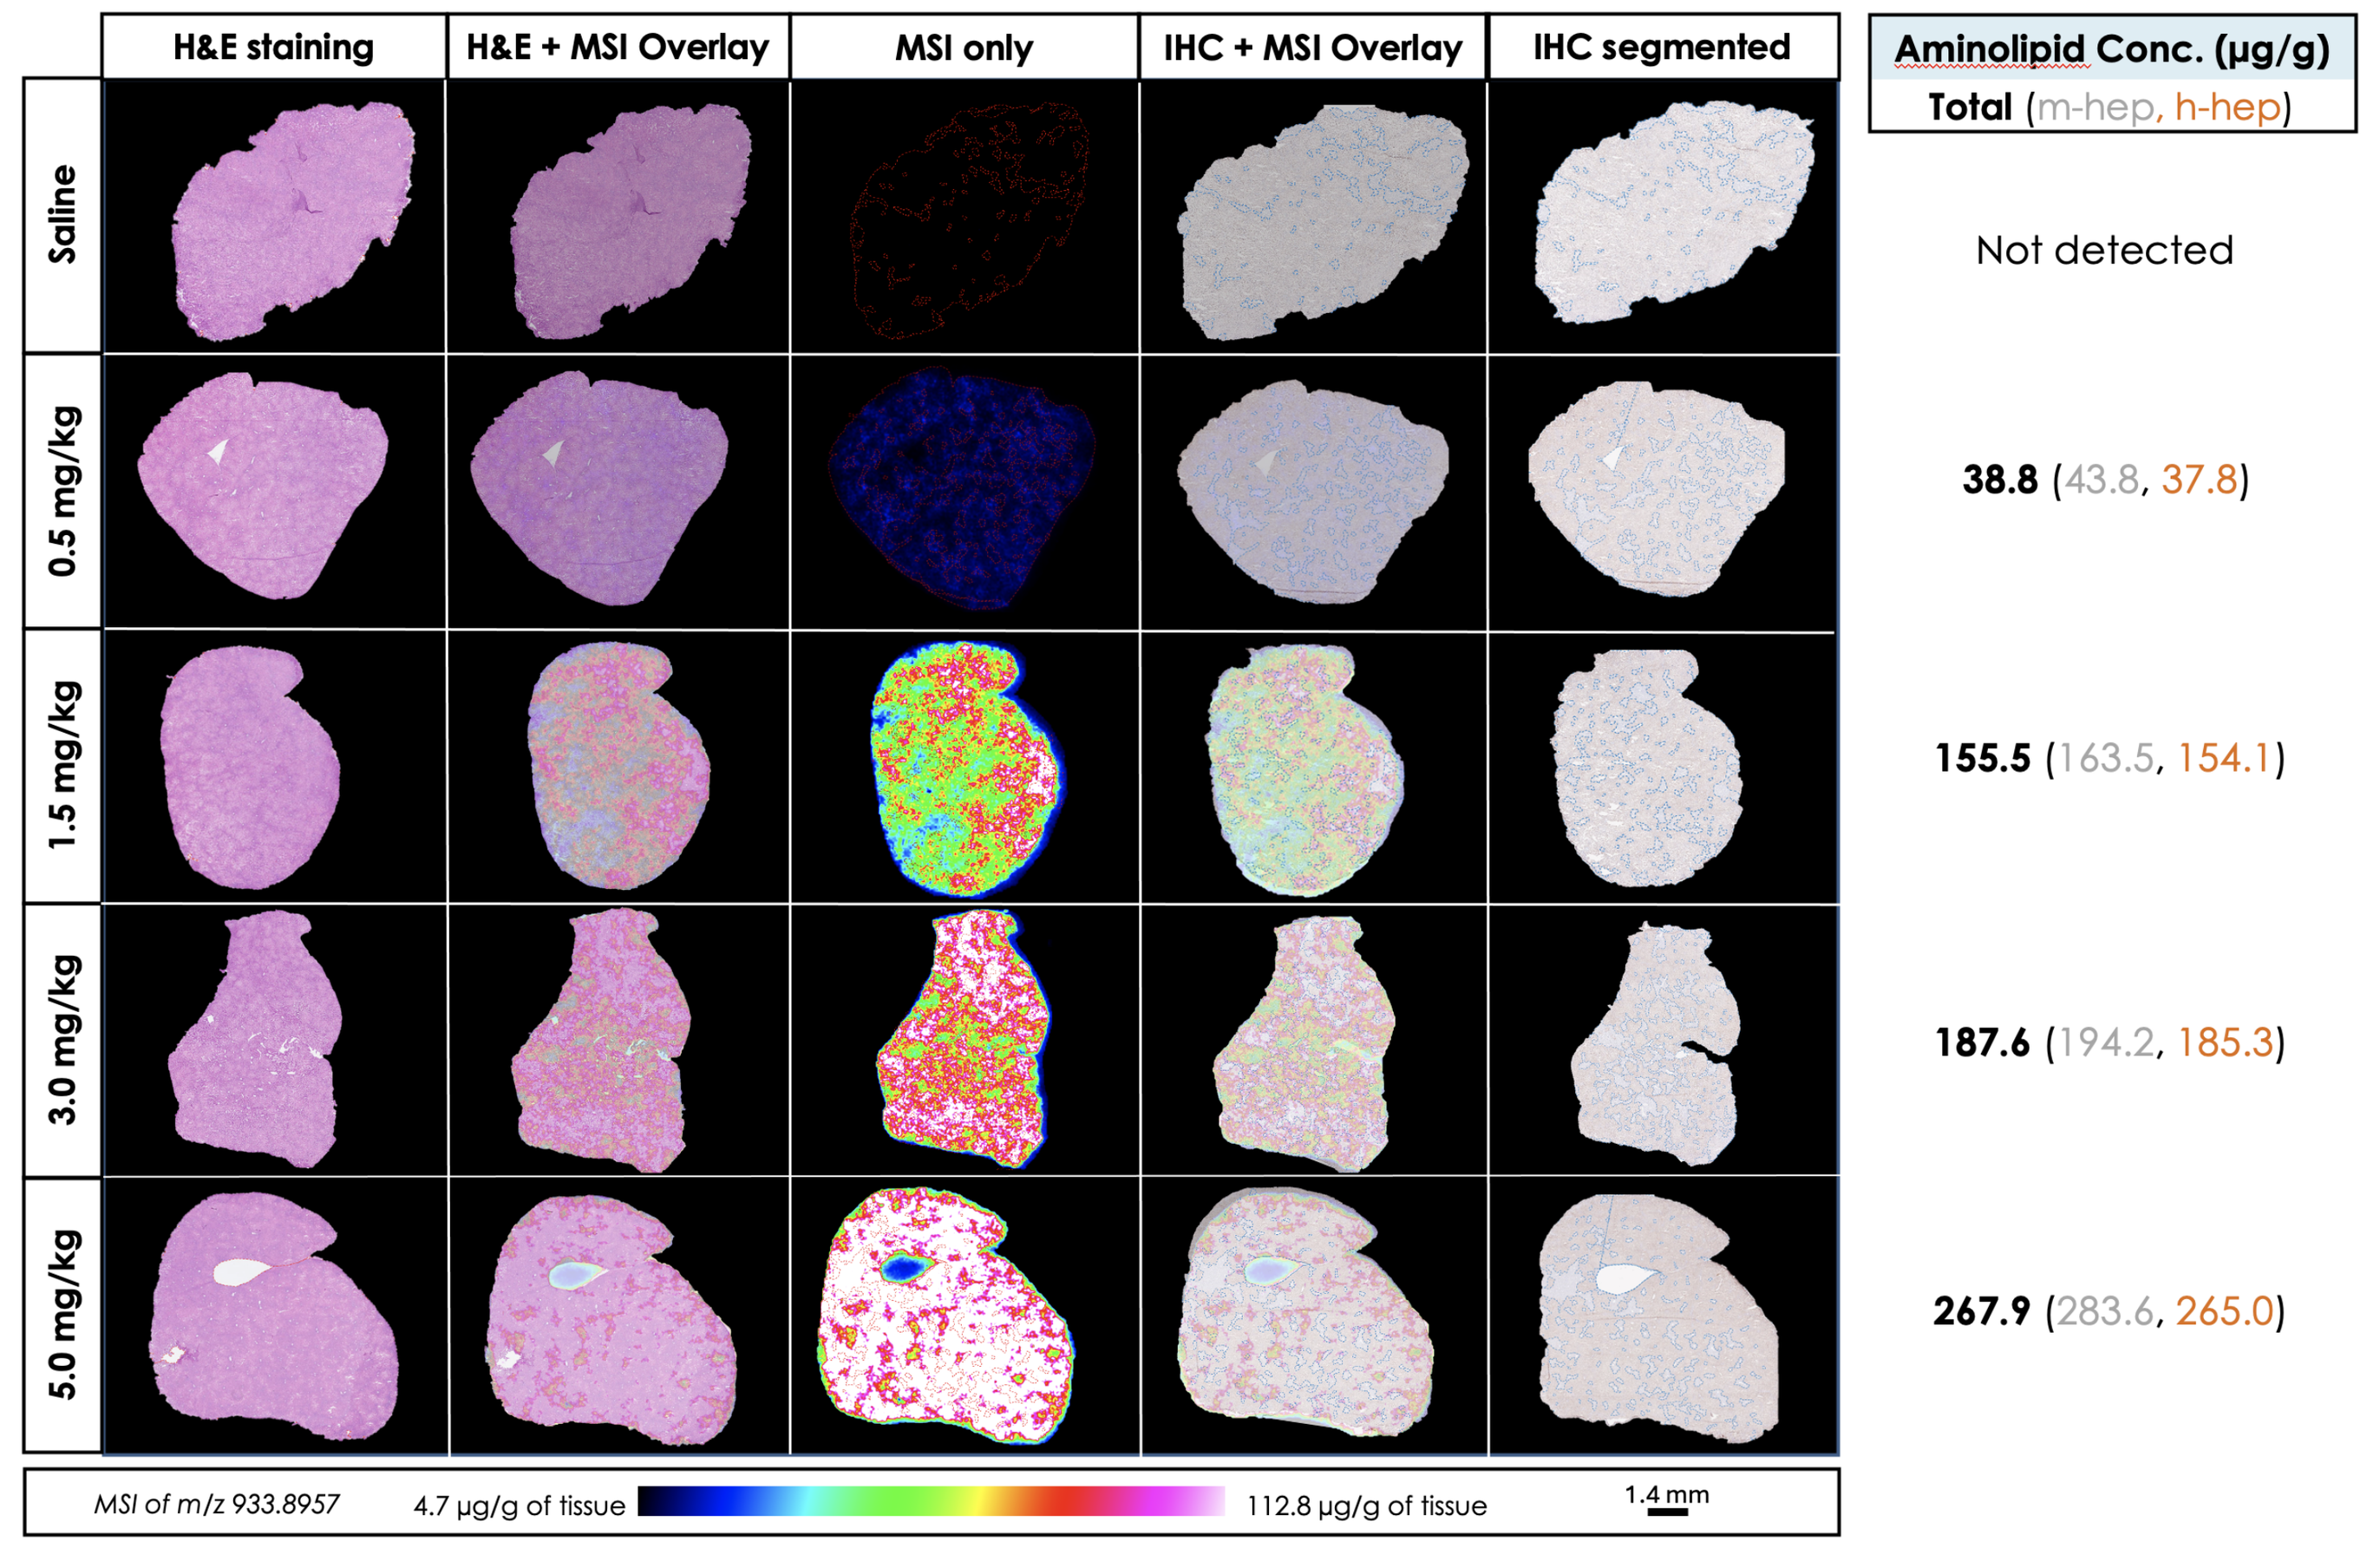
H&E staining
H&E + MSI Overlay
MSI only
IHC + MSI Overlay
IHC segmented
Saline
0.5 mg/kg
1.5 mg/kg
3.0 mg/kg
5.0 mg/kg
MSI of m/z 933.8957
4.7 µg/g of tissue
112.8 µg/g of tissue
1.4 mm

Aminolipid Conc. (µg/g) Total (m-hep, h-hep)
Not detected
38.8 (43.8, 37.8)
155.5 (163.5, 154.1)
187.6 (194.2, 185.3)
267.9 (283.6, 265.0)

Figure 4. Delayed protein expression in human hepatocytes suggests an impairment in mRNA-LNP uptake and/or translation

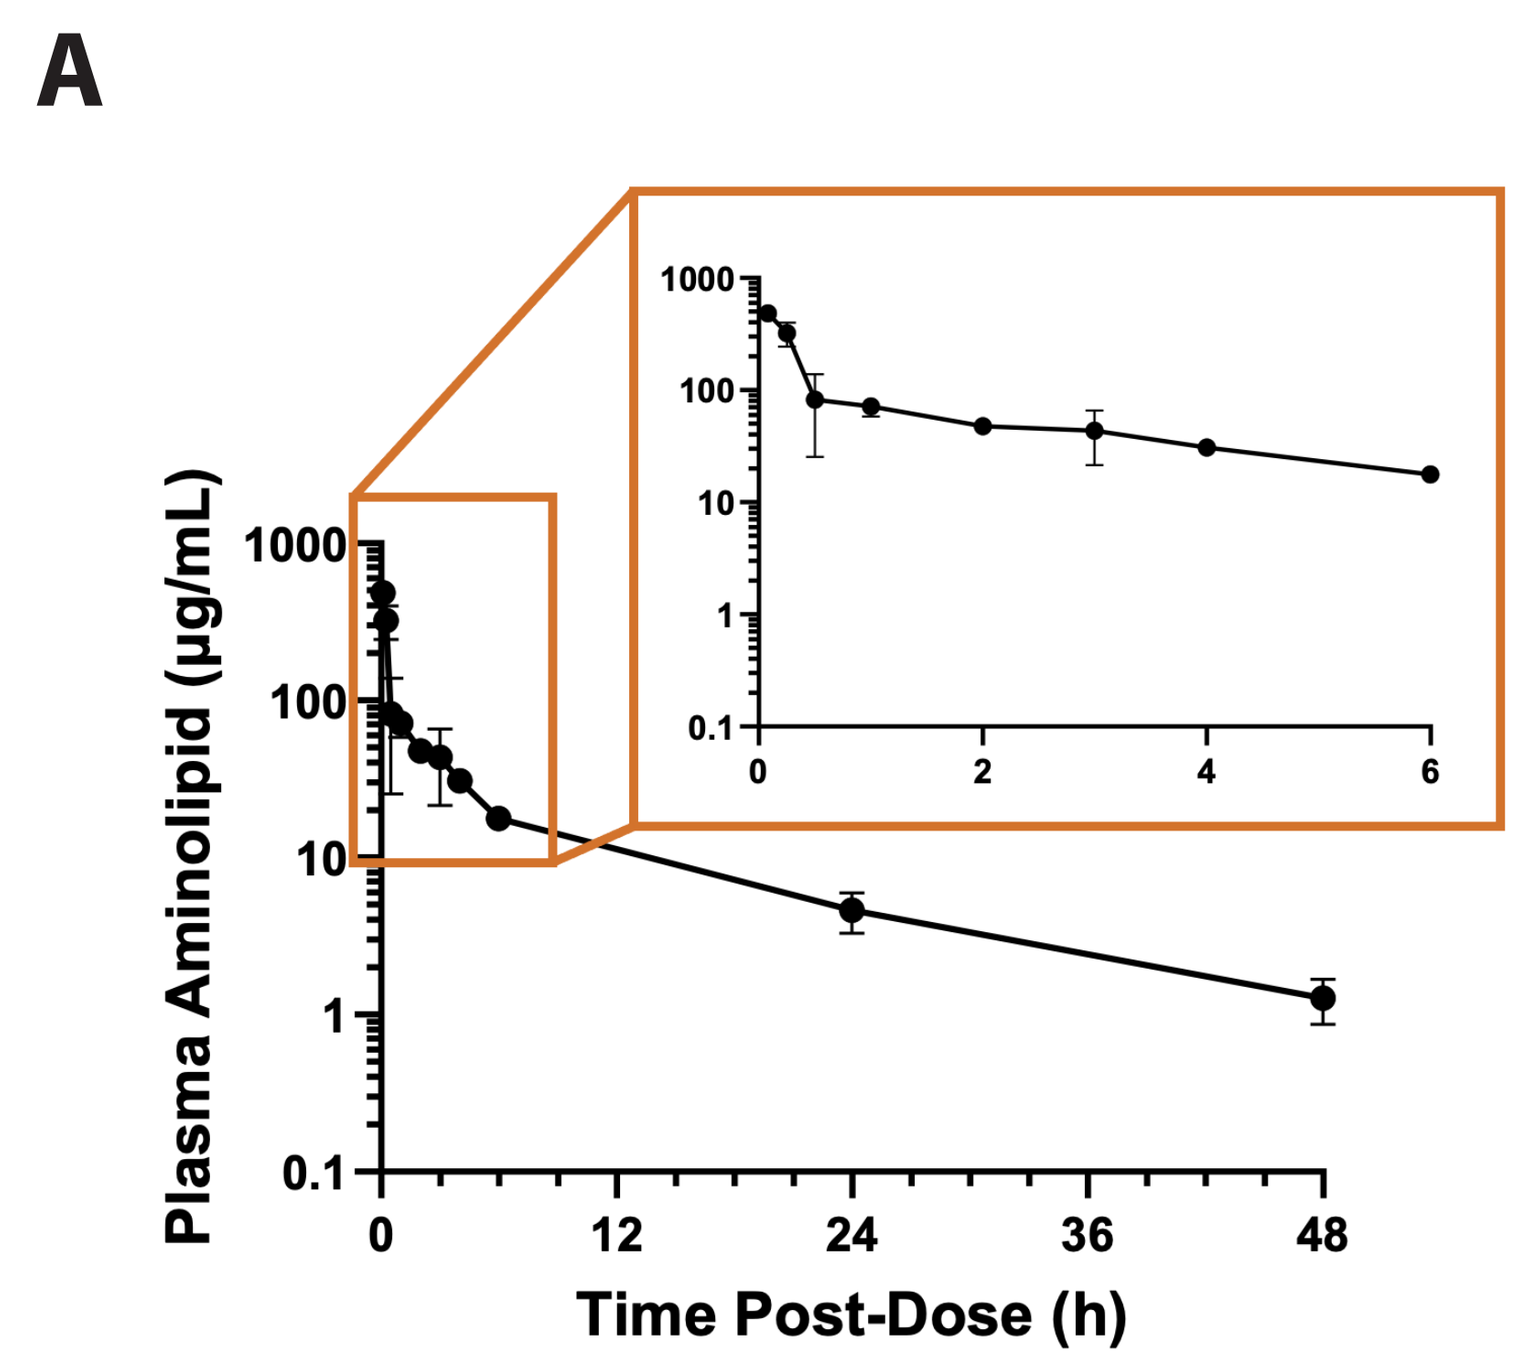
A
Plasma Aminolipid (µg/mL)
Time Post-Dose (h)

Pharmacokinetic study of LNP in PXB-mice. Mice were injected intravenously at 1.5 mg/kg with LNP13 containing IgG mRNA. Blood was sampled at different time points post-administration and processed to plasma for aminolipid quantification (A). Liver tissues were collected and processed for mRNA in situ hybridization (B) and immunohistochemistry (C). Plasma pharmacokinetic profile exhibited a rapid initial clearance of aminolipid from the blood compartment, followed by a slower terminal elimination phase.

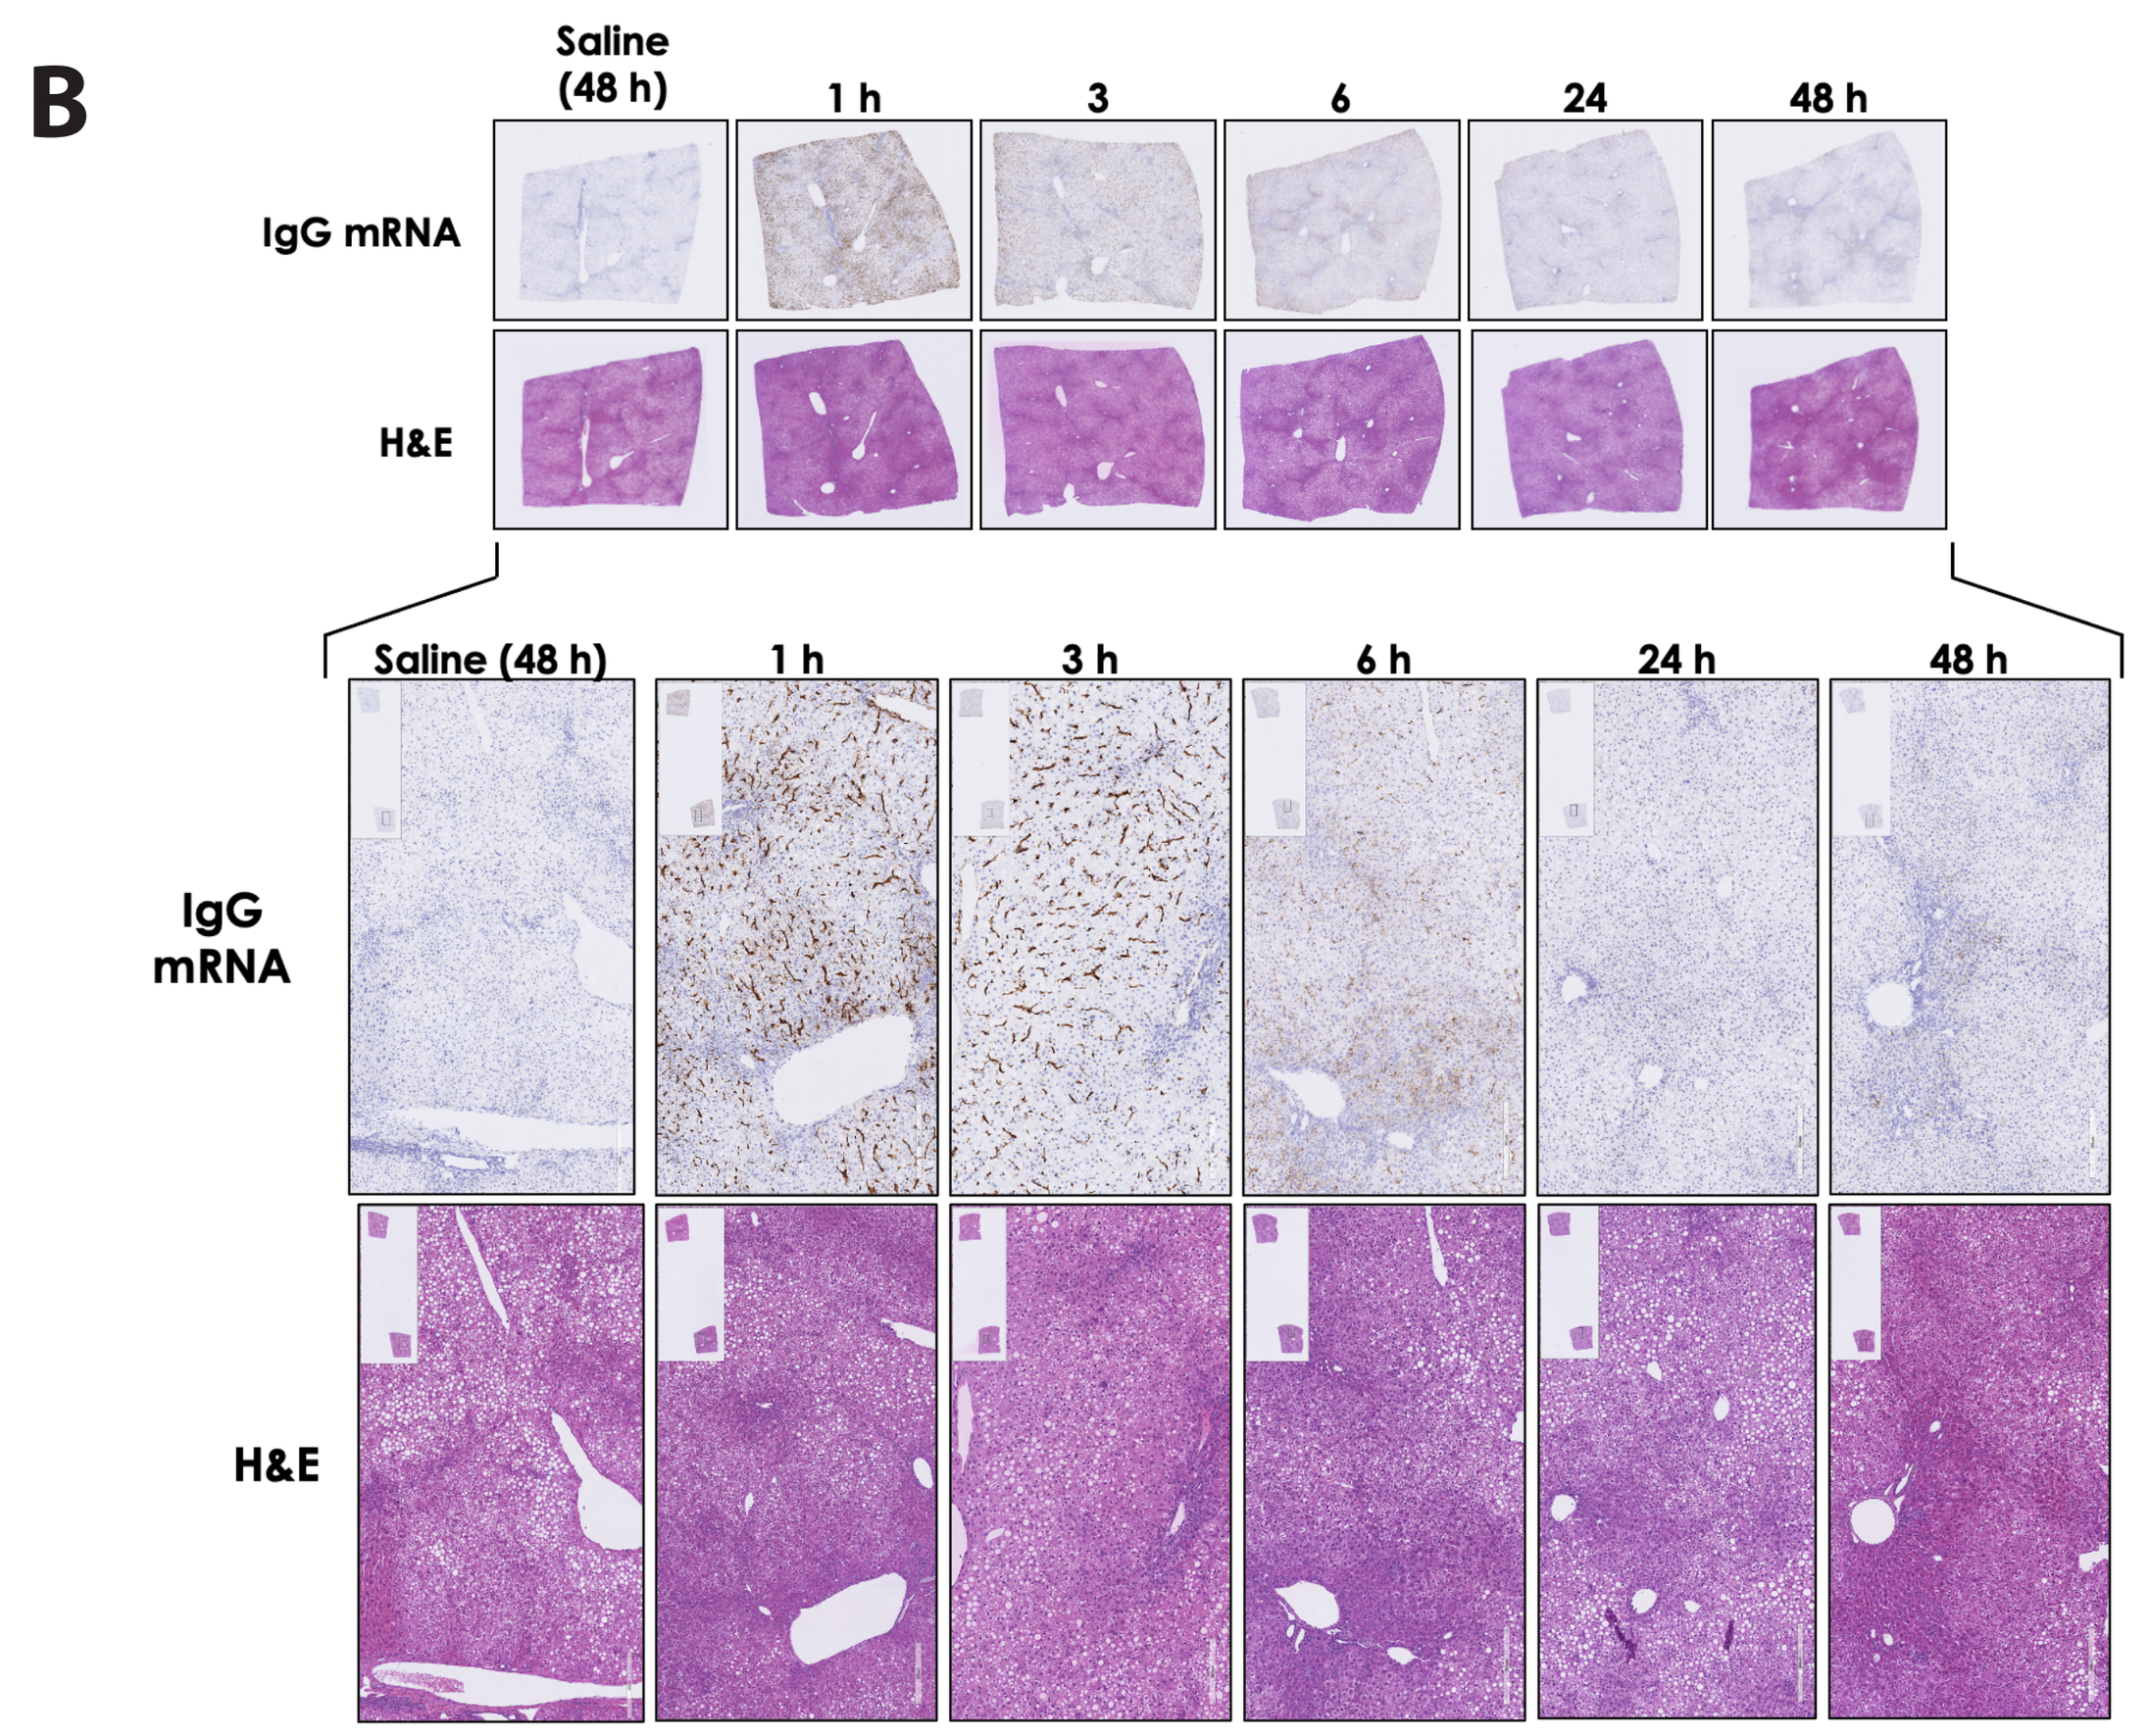
B
Saline (48 h)
1 h
3
6
24
48 h
IgG mRNA
H&E
Saline (48 h)
1 h
3 h
6 h
24 h
48 h

mRNA in situ hybridization (ISH). Serial liver sections were subjected to ISH using an IgG mRNA-specific probe (zoomed view, top) and H&E staining (zoomed view, bottom). ISH showed widespread distribution of payload throughout the liver at 1 h post-administration (macro view).

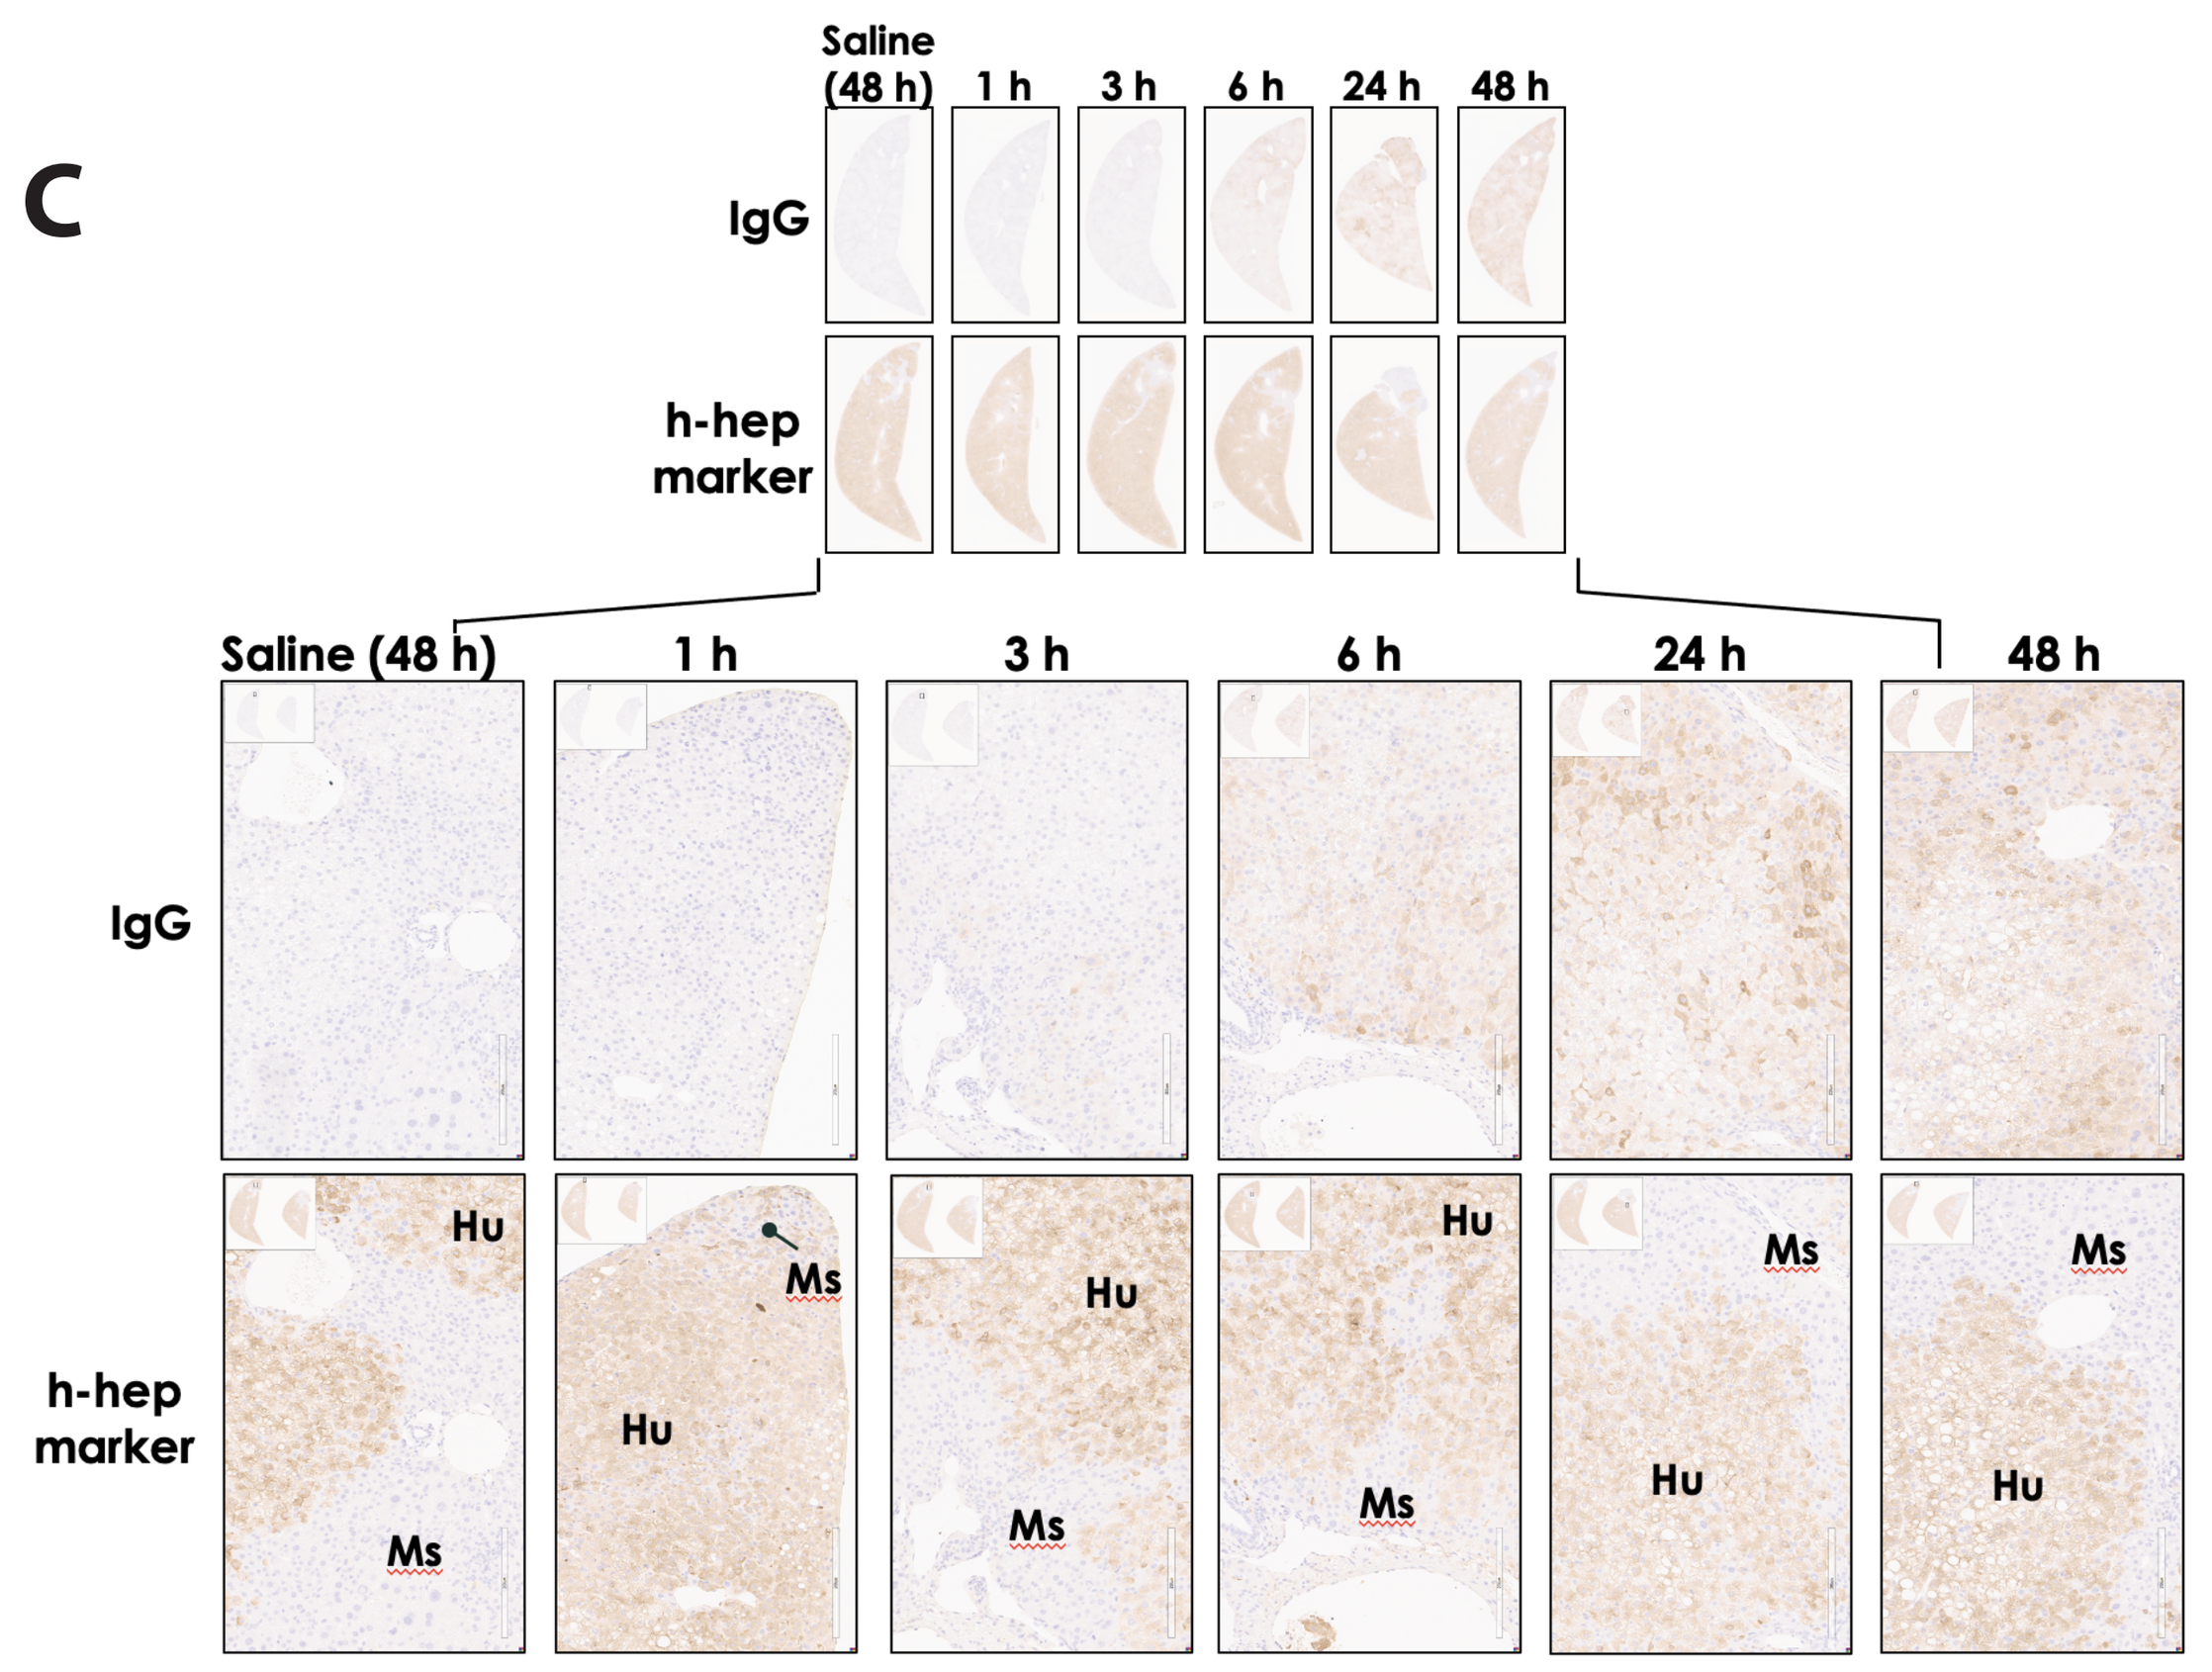
C
Saline (48 h)
1 h
3 h
6 h
24 h
48 h
IgG
h-hep marker
Hu
Ms

Immunohistochemistry staining of LNP13-treated mice livers. Serial liver sections were stained using antibodies specific to IgG protein (zoomed view, top) and a human hepatocyte-specific marker (zoomed view, bottom). IgG protein expression was observed as early as 3 h post-administration and was more robust in murine hepatocytes. At later time points (24 to 48 h), this differential in expression relative to human hepatocytes appeared to be reduced. These data altogether suggest a potential delay in the uptake and/or expression of mRNA-LNP in human hepatocytes. Hu = Human hepatocytes; Ms = Mouse hepatocytes

Summary

1. mRNA-LNP exhibited varying potencies following systemic administration in PXB-mice, with a relative rank order of LNP13>LNP07>LNP09. Despite dose-related increases in liver transaminases, mRNA-LNP were generally well tolerated (up to 5.0 mg/kg for LNP13), with no observed clinical signs at moderately high dose levels.
2. QMSI revealed extensive distribution of LNP-derived aminolipid in the liver, with no preferential delivery to murine or human hepatocytes. mRNA in situ hybridization showed similar, widespread distribution of the payload at early time points post-administration.
3. Delayed protein expression in human hepatocytes suggests a potential impairment in uptake and/or translation compared to murine hepatocytes.

References

1. Barry, J. et al. Multicenter Validation Study of Quantitative Imaging Mass Spectrometry. Analytical Chemistry (2019).

Acknowledgements

We thank PhoenixBio for the provision of PXB-mice and supporting the in-life phase of these studies.