Improved T Cell Delivery and Expression of mRNA Using CD8-Targeted Lipid Nanoparticles with Athebody® Designed Ankyrin Repeat Proteins (DARPins)

Fan Yan[1], Sherry A. Weppler[1], Thomas Chamberlain[1], Grayson Wong[1], Ghania Chikh[1], Nora Guidotti[2], Raffaella Capozzoli[2], Julien Hiblot[2], Rohan Eapen[2], Ying K. Tam[1], Barbara L. Mui[1]

1 Acuitas Therapeutics, Vancouver, BC, Canada. | 2 Athebio AG, Zürich, Switzerland.

Non-targeted LNP activity in mouse and human immune cells

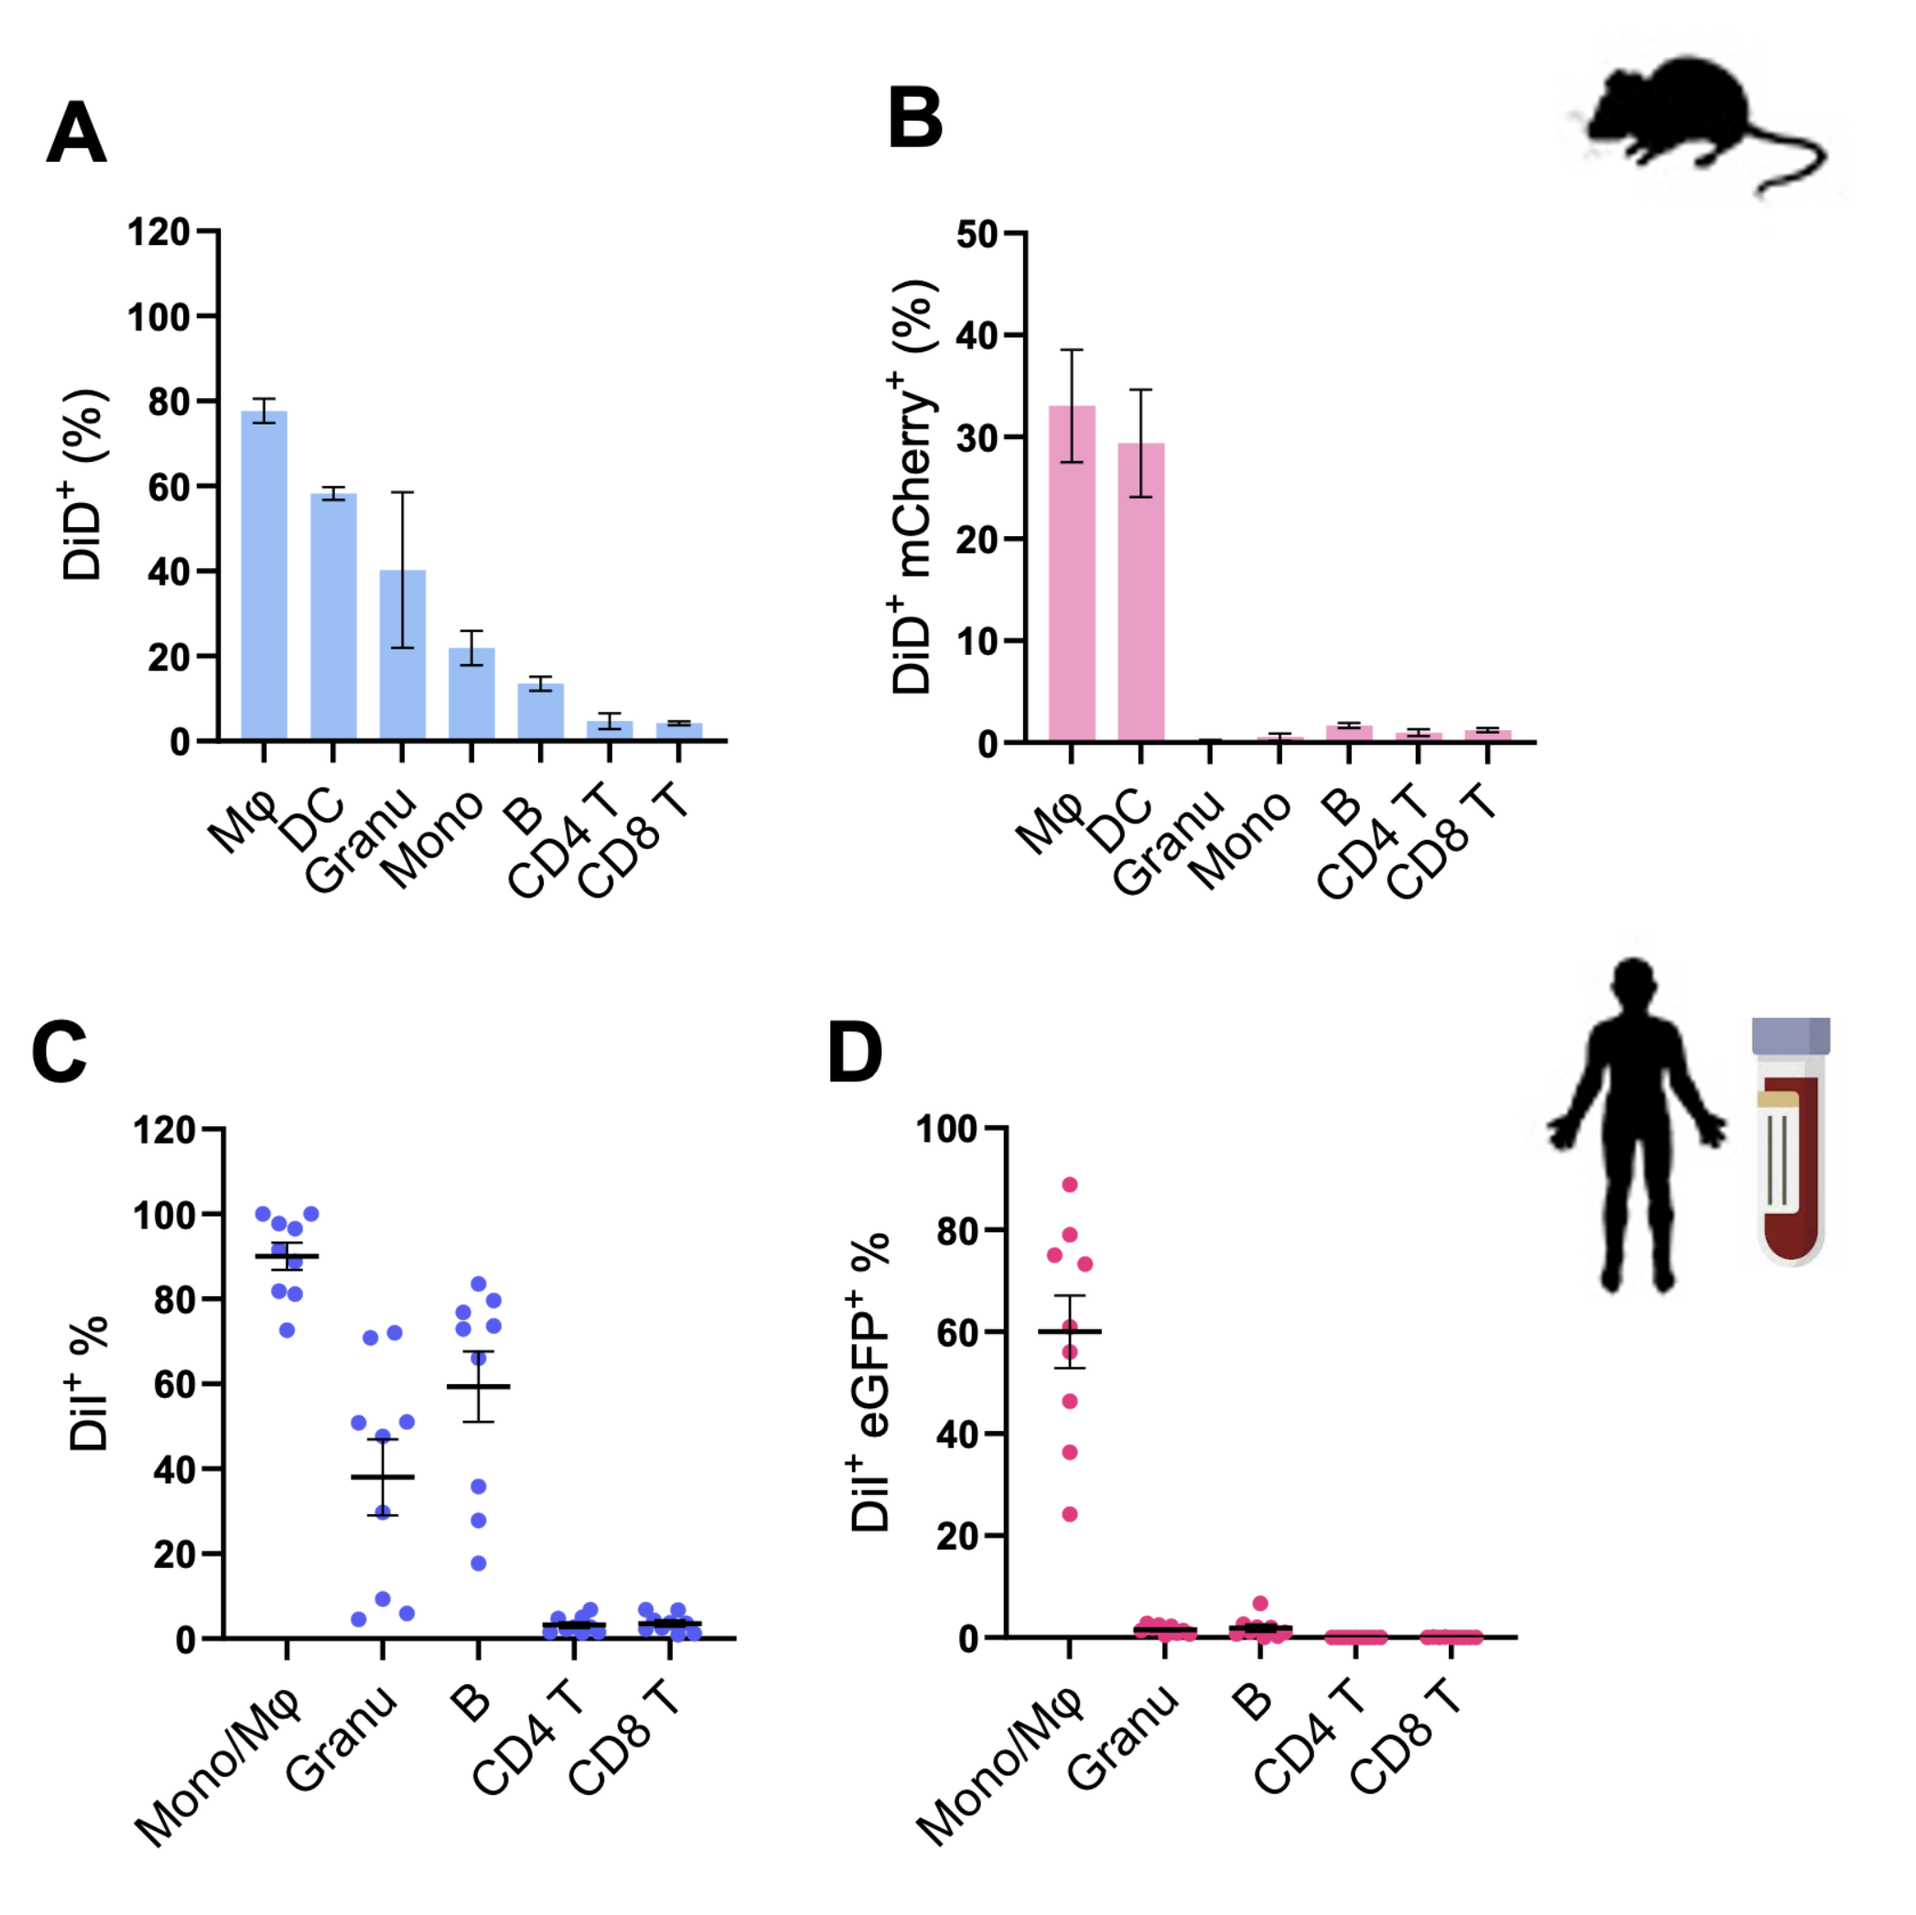
A
DiD⁺ (%)
Mφ DC Granu Mono B CD4 T CD8 T
B
DiD⁺ mCherry⁺ (%)
C
DiI⁺ %
Mono/Mφ Granu B CD4 T CD8 T
D
DiI⁺ eGFP⁺ %

Figure 1. Characterizing cellular distribution of non-targeted (native) LNP.
% of LNP binding/uptake (**A**) and RNA expression (**B**) of native LNP in mouse splenocytes. % of binding/uptake (**C**) and expression (**D**) of native LNP in human blood cells.

Dose range finding and DARPin:LNP ratio titration for DARPin-LNP

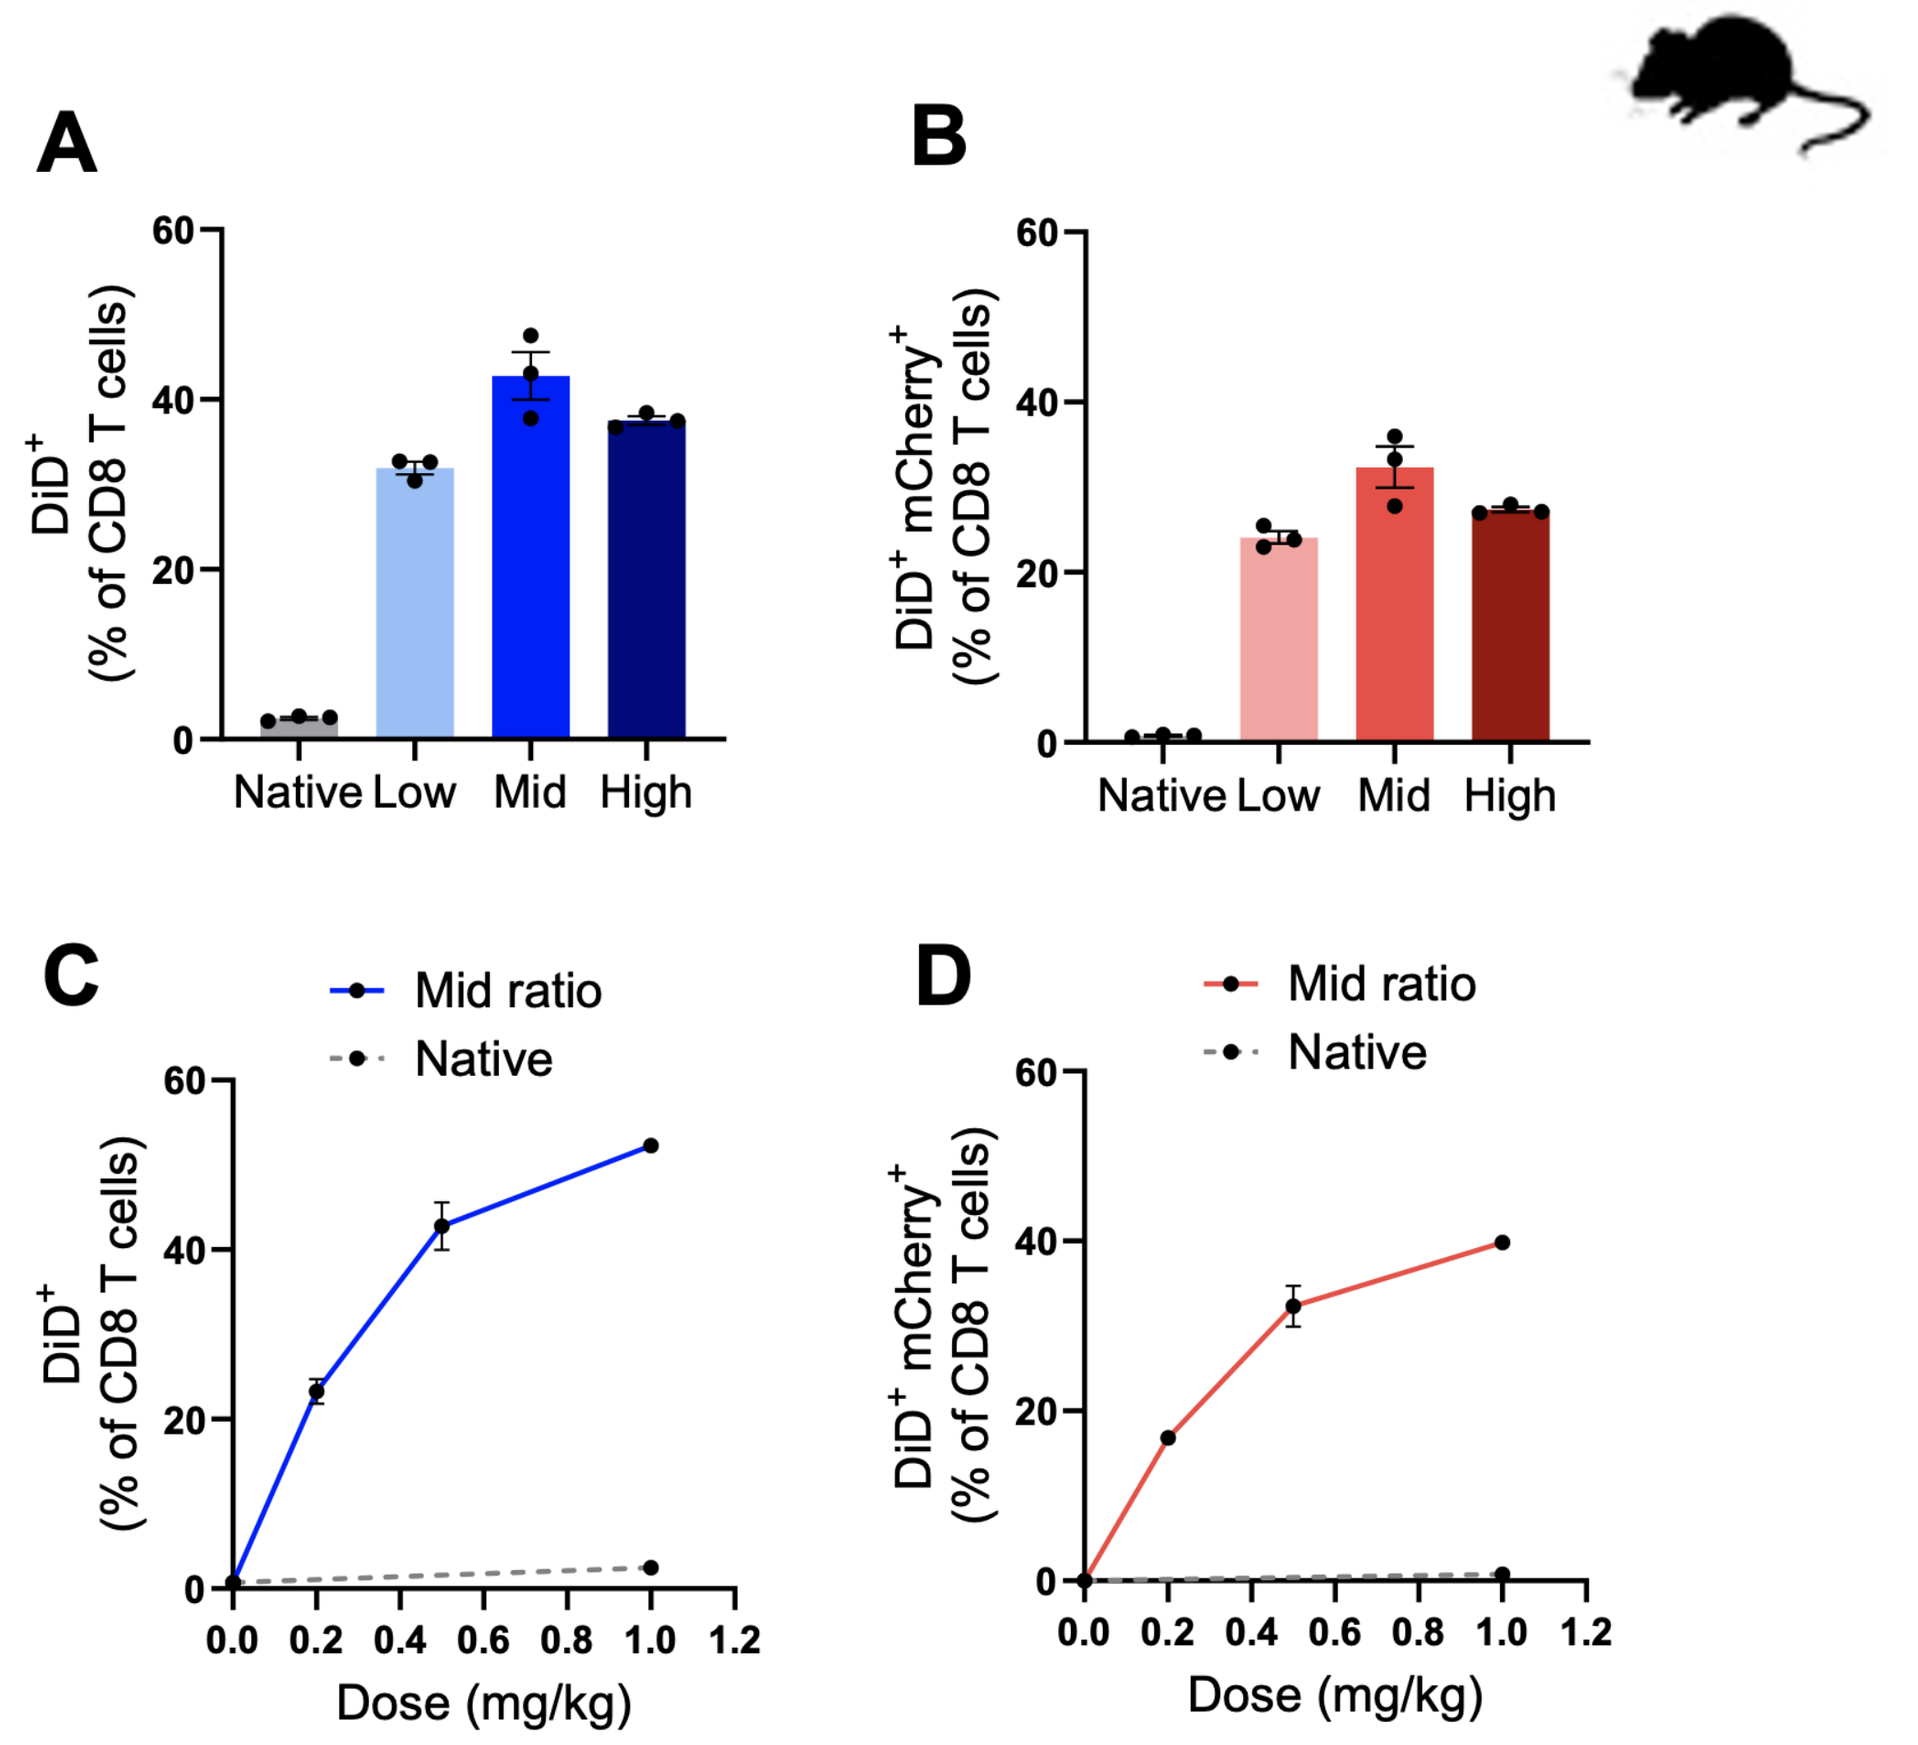
A
DiD⁺ (% of CD8 T cells)
Native Low Mid High
B
DiD⁺ mCherry⁺ (% of CD8 T cells)
C
Mid ratio
Native
Dose (mg/kg)
D

Figure 2. Dose and ratio titration of DARPin-LNP.
% of binding/uptake (A) and expression (B) of formulations at various DARPin:LNP ratios in CD8 T cells. Dose-effect curves for binding (C) and expression (D) of LNP in CD8 T cells.

CD8 DARPin screen and DARPin conjugation

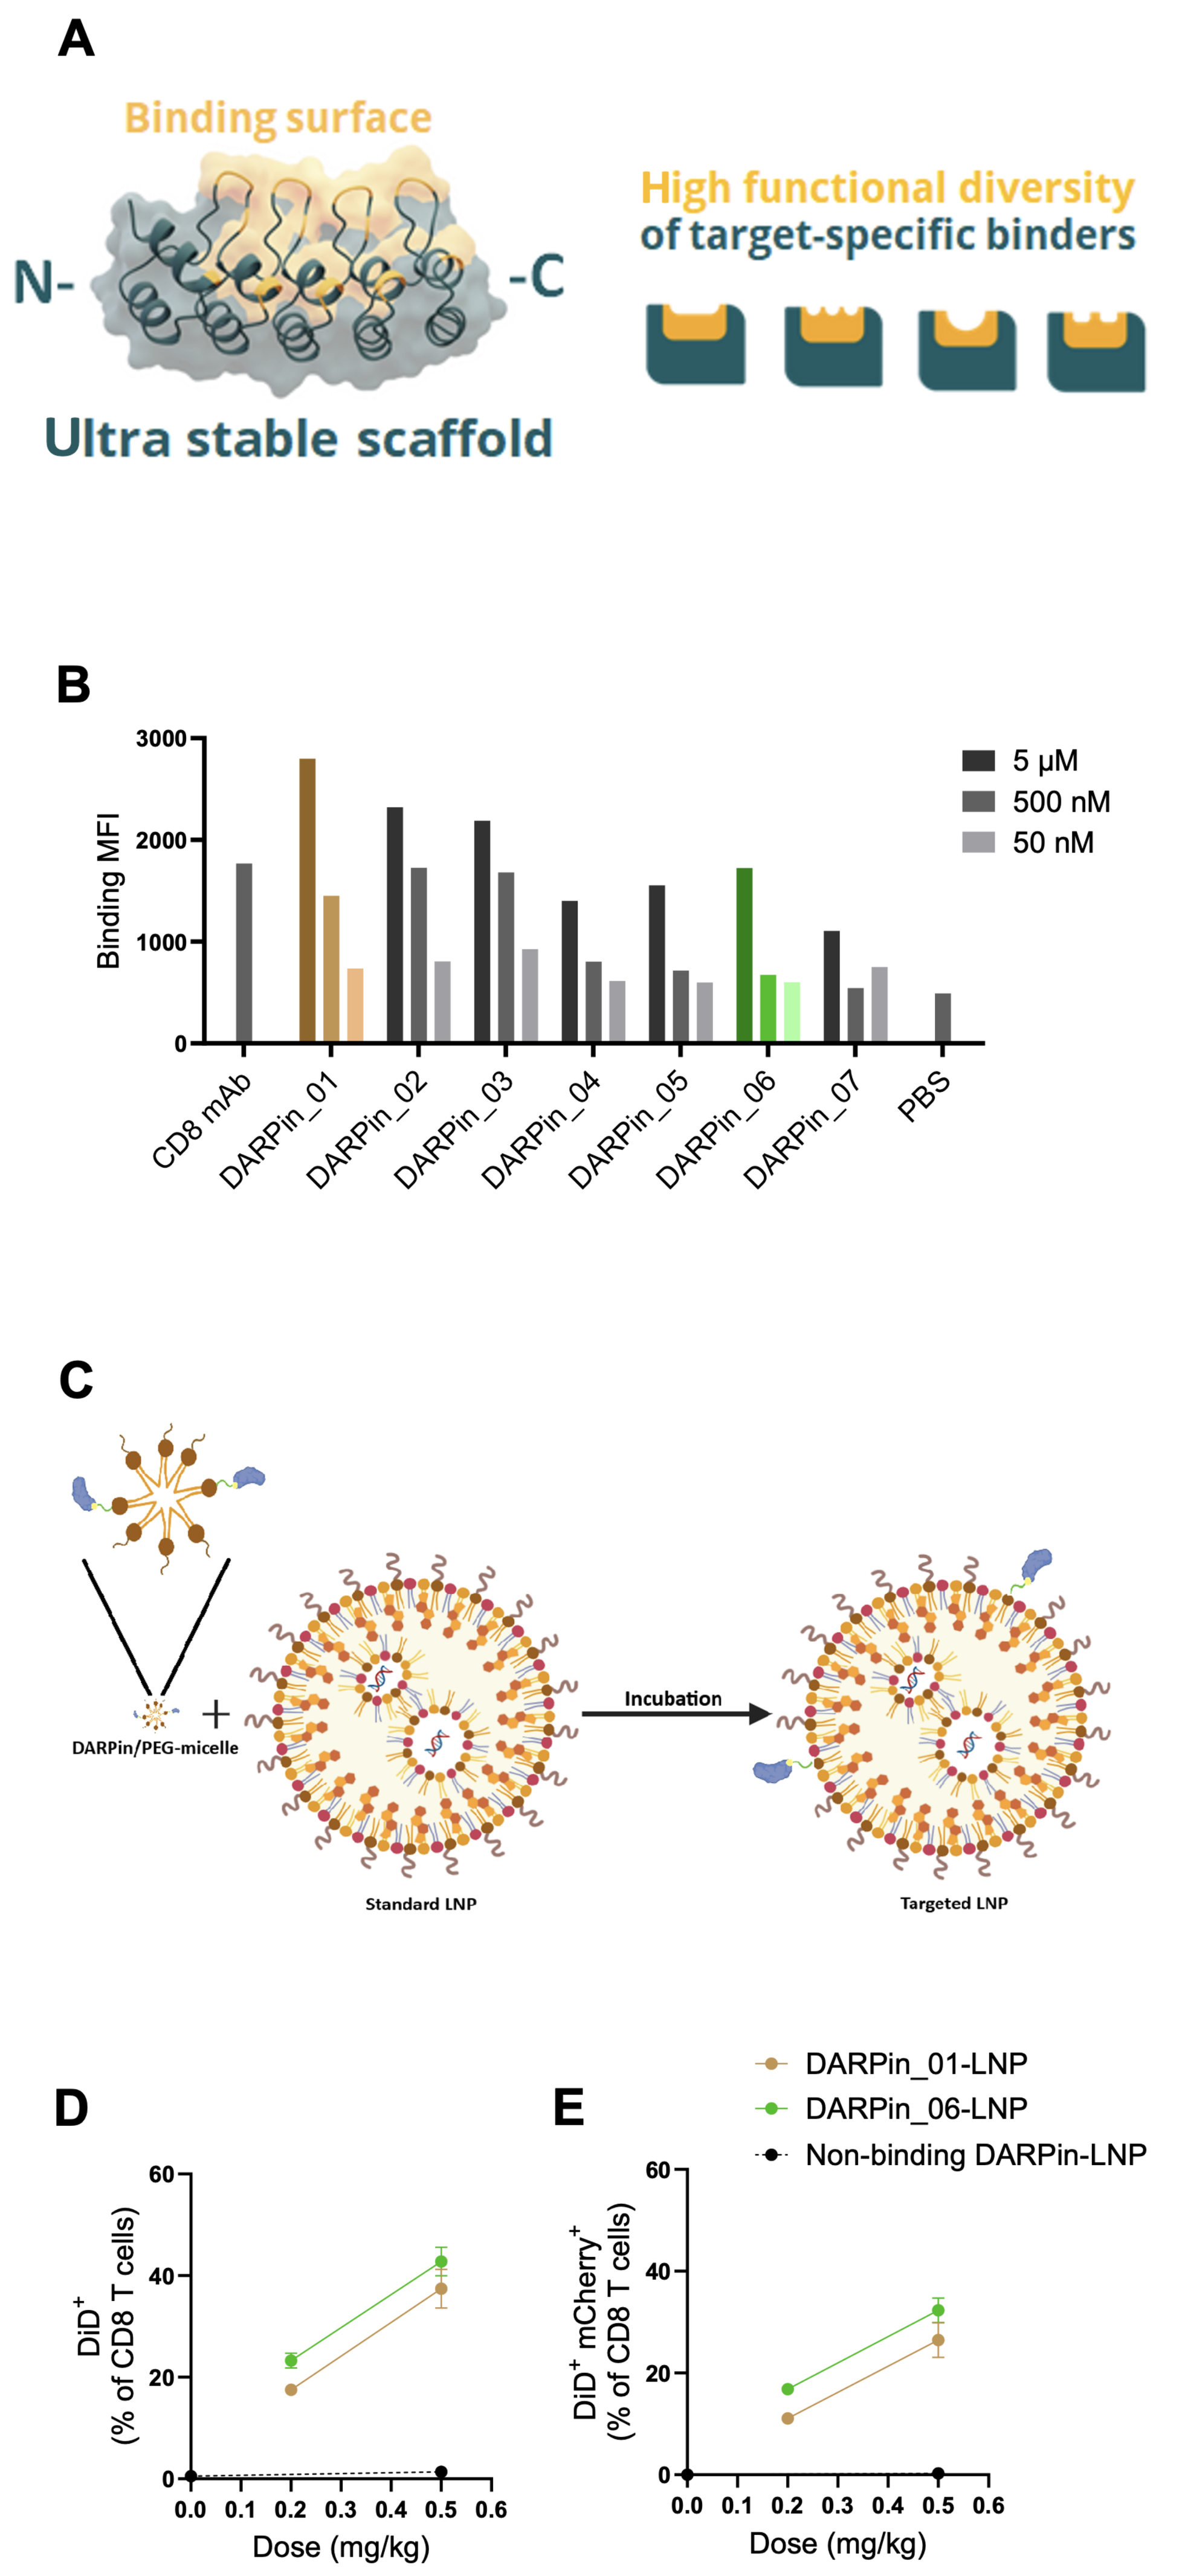
A
Binding surface
N-
-C
Ultra stable scaffold
High functional diversity of target-specific binders
B
Binding MFI
5 µM
500 nM
50 nM
CD8 mAb DARPin_01 DARPin_02 DARPin_03 DARPin_04 DARPin_05 DARPin_06 DARPin_07 PBS
C
DARPin/PEG-micelle
Incubation
Standard LNP
Targeted LNP
D
E
DARPin_01-LNP
DARPin_06-LNP
Non-binding DARPin-LNP
DiD⁺ (% of CD8 T cells)
DiD⁺ mCherry⁺ (% of CD8 T cells)
Dose (mg/kg)

Figure 3. DARPin selection and conjugation to LNP.
A: DARPin structure. **B**: Screen of DARPin candidates by binding MFI to CD8 T cells. **C**: DARPin conjugation to LNP. **D**, **E**: LNP-conjugated DARPin selection in mice.

Abstract

Lipid nanoparticles (LNP), administered intravenously, primarily accumulate in tissues with fenestrated vasculature (e.g., liver, spleen, bone marrow) due to vascular permeability. In the liver, endogenous apolipoprotein E (apoE) binds to the LNP surface, mediating hepatocyte uptake and nucleic acid payload delivery via low-density lipoprotein receptors. While LNP delivery to the liver is well established, expanding delivery to extrahepatic tissues remains a challenge. One approach is "active" targeting, where ligand-conjugated LNP facilitate direct uptake by target cells in the blood compartment. Here, we evaluate Athebody® designed ankyrin repeat proteins (DARPins), antibody mimetics generated through synthetic combinatorial libraries and in vitro and in silico selection methods, as high affinity and specificity ligands for targeted LNP delivery.

Using DiD-labeled LNP containing a fluorophore-expressing mRNA, we first evaluated binding and fluorescent protein expression in mouse splenocytes after IV injection of non-targeted LNPs. At 24 hours after administration, splenic macrophages (33%) and dendritic cells (29%) exhibited notable LNP uptake and payload mRNA expression, while granulocytes, B cells, and T cells showed minimal engagement (<2%). Similar trends were observed in vitro using human whole blood. To enable T cell targeting, we conjugated a murine CD8-targeting DARPin to LNPs, and injected these IV into mice to assess in vivo targeting efficiency. CD8 DARPin-targeted mRNA-LNP showed dose-dependent, target-specific binding (44%) and transgene expression (34%) in CD8 T cells, with no impact on viability (>90%). Using LNPs with extended circulation, in vivo targeting efficiency increased to 89% binding and 59% expression in CD8 T cells, with additional uptake in CD8⁺ dendritic cells (88% binding, 61% expression). There was no significant increase in targeting to CD8⁻ cells (CD8⁻ DCs, CD4 T cells, B cells, or myeloid cells) compared to unconjugated LNPs. Translating this to human cells, a human CD8-targeting DARPin-LNP showed ~98% binding and 46–90% expression in CD8 T cells from healthy human donor whole blood. These findings highlight DARPin-conjugated LNPs as a promising strategy for highly specific mRNA delivery to hard-to-transfect cells accessible within the vascular compartment.

Results

Non-targeted LNP activity in murine and human immune cells

- Extrahepatic LNP delivery to major immune cell types was evaluated for both LNP binding/uptake (DiD+ or DiI+) and RNA expression (mCherry+ or eGFP+). Mice were injected IV with non-targeted (native) LNP. After 24 hours, spleens were collected and splenocytes were analyzed by flow cytometry. LNP binding/uptake was observed in macrophages (Mφ), DC, granulocytes, monocytes and B cells in mice. However, only Mφ (33%) and DC (29%) demonstrated mRNA expression, while the other cell types did not (<2%) (Fig. 1A & B).
- For human cells, whole blood was collected from healthy donors, and incubated with 0.5 µg/mL native LNP for 24 hours. Similarly, monocytes/Mφ, granulocytes and B cells showed notable LNP binding/uptake but only monocytes/Mφ elicited RNA expression (60%) (Fig. 1C & D).
- T cells showed only minimal binding/uptake and expression following treatment with native LNP.

CD8 DARPin screen and DARPin conjugation

- Designed ankyrin repeat proteins (DARPins) are a class of antibody mimetics composed of a N-and C-cap and 2-4 library modules that contain the variable (target binding) region. Notably, these DARPins contain only one conjugation site that allows site-specific conjugation and ensures a preferred orientation of the DARPin on the LNP (Fig. 3A).
- A set of 7 DARPins were pre-screened by their affinity to CD8 extracellular domain and good stability (data not shown). The binding/uptake capability to splenic CD8 T cells was assessed (Fig. 3B). According to binding MFI to ex vivo CD8 T cells, DARPin_01 (strong binder) and DARPin_06 (intermediate binder with improved stability) were selected to proceed screening with LNP. DARPins were conjugated to PEG-lipid micelles and then incubated with standard LNP to allow post insertion, forming targeted LNP (Fig. 3C).
- DARPin-conjugated LNP were administered IV to mice. After 24 hours, both DARPin_01 and DARPin_06 efficiently directed LNP to CD8 T cells indicated by significant binding/uptake and expression. Although DARPin_06 showed lower binding/uptake ability in DARPin screening, DARPin_06-LNP performed slightly better than DARPin_01-LNP in vivo, possibly due to its improved stability (Fig. 3D).

Dose range finding and DARPin:LNP ratio titration for DARPin-LNP

- DARPin-LNP were formulated at low, medium and high DARPin_06:LNP ratios, individually administrated to mice at 0.2, 0.5, 1.0 mg/kg. All DARPin-LNP induced target-specific binding/uptake and expression compared to native LNP. Medium ratio induced the best LNP activity in CD8 T cells (Fig. 2A & B).
- Dose-activity curves indicated DARPin-LNP (mid-ratio) had dose dependent activity from 0.2 to 1.0 mg/kg (Fig. 2C & D). 0.5 mg/kg was selected for the following mouse studies.

Activity comparison with mAb-LNP

- A commercially available CD8 mAb (clone 53-6.7) was conjugated to LNP and compared side-by-side with DARPin-LNP. Both mAb and DARPin-LNP can elicit targeted binding/uptake and expression in CD8 T cells (Fig. 4A & B). The overall activity of DARPin-LNP was significantly better than mAb-LNP while the viability (data not shown) and activity in other cell types were not significantly affected (Fig. 4D & E).
- No cage-side observations were noted for DARPin-LNP, but reduced activity and piloerection were seen with mAb-LNP animals at 0.5 mg/kg (Fig. 4C).

Improved targeted activity by long-circulating LNP

- To further enhance the extrahepatic delivery of DARPin-LNP, LNP composition was optimized to prolong circulation lifetime and reduce liver distribution. We incorporated a PEG-lipid species which is retained with the LNP and successfully elevated the targeted activity (data not shown).
- However, given the potential desire for repeat dosing where a persistent PEG may be associated with development of undesired adaptive immunity, we made compositional alterations without changing PEG-lipid to extend circulation lifetime. Four long-circulating variants were designed, produced and compared in the DARPin-LNP format. V1-4 were all able to improve LNP binding/uptake and RNA expression compared to the standard composition. V3 gave a superior activity at 89% binding/uptake and 59% expression which improved MFI by 5.7 and 4.1 folds compared to standard LNP (Fig. 5 A-D).
- We also assessed LNP distribution and activity in liver. Compared to standard LNP, most variants showed less distribution to the liver, while all mediated large reductions in RNA expression (Fig. 5E & F), suggesting that decreased hepatic delivery may contribute to better targeted delivery and activity.

Storage stability of DARPin-LNP

- Particle characteristics and targeted activity (in SUP-T1 T cell line) were monitored at multiple time points after storage at -80°C for DARPin-conjugated LNP with different cationic lipids (LNP-07, 09 and 13). The mRNA encapsulation was consistently good for all 3 cationic lipid-based DARPin-LNP (>90%), with no loss observed even at later time points (Fig. 6A). Similarly, LNP size varied slightly at different time points but stayed within a tight range (Fig. 6B) while LNP polydispersity index (PDI) of LNP-07 and 13 was consistent but more instability observed with LNP-09 (Fig. 6C). In vitro, there were no consistent decreases in LNP binding/uptake (Fig. 6D) and reporter gene expression (Fig. 6E). Overall, these data demonstrate stable activity after long storage up to 7 months with better performance for LNP-07 and 13 compared to LNP-09.

CD8 targeted binding/uptake and activity in human blood

- Two DARPins against human CD8 were designed and validated. Human CD8 DARPin-LNP were made and added at 0.5 µg/mL concentration to human whole blood culture to evaluate binding and expression. Nearly complete binding/uptake (>98%) and a high portion of reporter RNA expression (40-69%) were observed in CD8 T cells at 24 hours (Fig. 6D), providing further evidence for clinical application.

Summary

- We have demonstrated promising extrahepatic delivery capability of LNP, by successful development of proprietary DARPin-conjugated LNP.
- A body of validation and optimization work with murine in vivo model and human samples has proved that DARPin-LNP can induce a substantial level of binding/uptake and expression in target cells without impact on cell viability. Long circulating LNP variants can further augment T cell delivery and reduce hepatic distribution and activity.
- Superior activity and better animal tolerability of DARPin-LNP was shown over mAb-LNP, likely due to the smaller molecule size, no Fc region and controllable conjugating orientation of DARPins.
- Long-term storage at -80°C for 6~7 months didn't significantly impact the LNP characteristics and activity, providing benefit for clinical usage.

Activity comparison with mAb-LNP

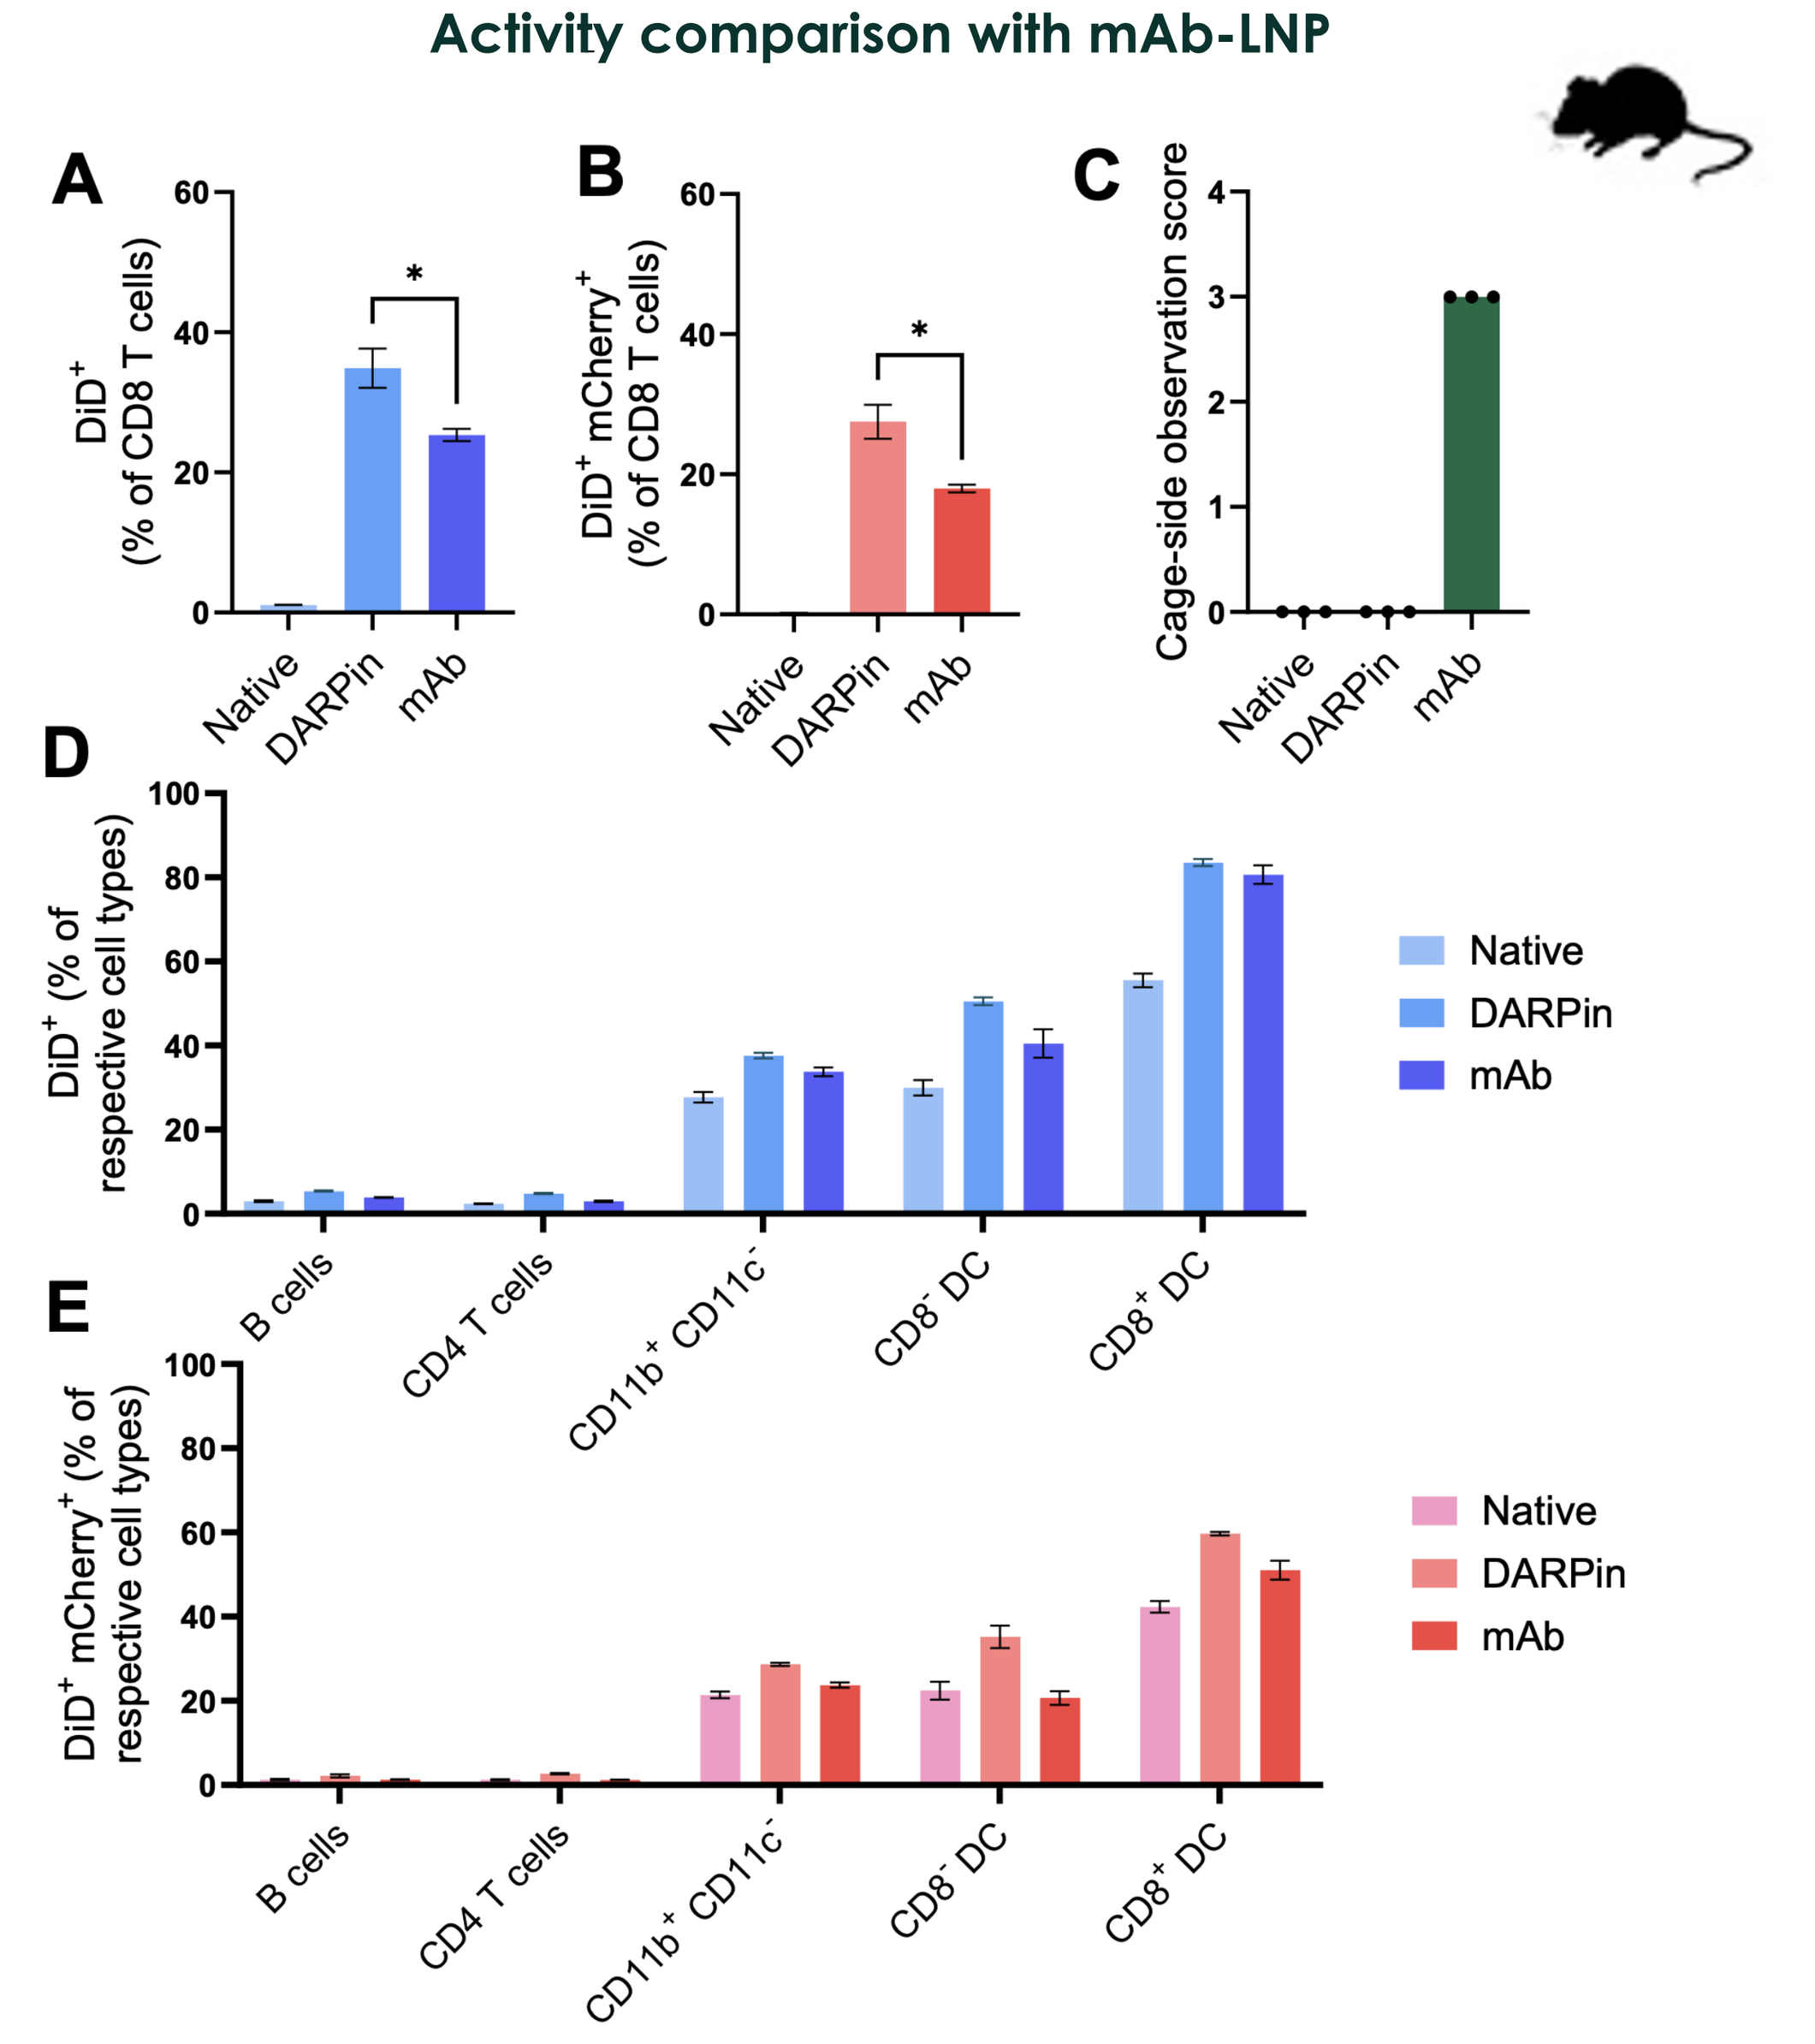
A
DiD⁺ (% of CD8 T cells)
Native DARPin mAb
B
DiD⁺ mCherry⁺ (% of CD8 T cells)
C
Cage-side observation score
D
DiD⁺ (% of respective cell types)
Native
DARPin
mAb
B cells CD4 T cells CD11b⁺ CD11c⁻ CD8⁻ DC CD8⁺ DC
E
DiD⁺ mCherry⁺ (% of respective cell types)

Figure 4. DARPin-LNP activity comparison to mAb-LNP
% of LNP binding/uptake (**A**), RNA expression (**B**) in splenic CD8 T cells and cage-side observation score (**C**) after administration of t-LNPs. % of binding/uptake (**D**) and expression (**E**) of other cell types after administration of t-LNP.

Improve targeted activity by long-circulating LNP

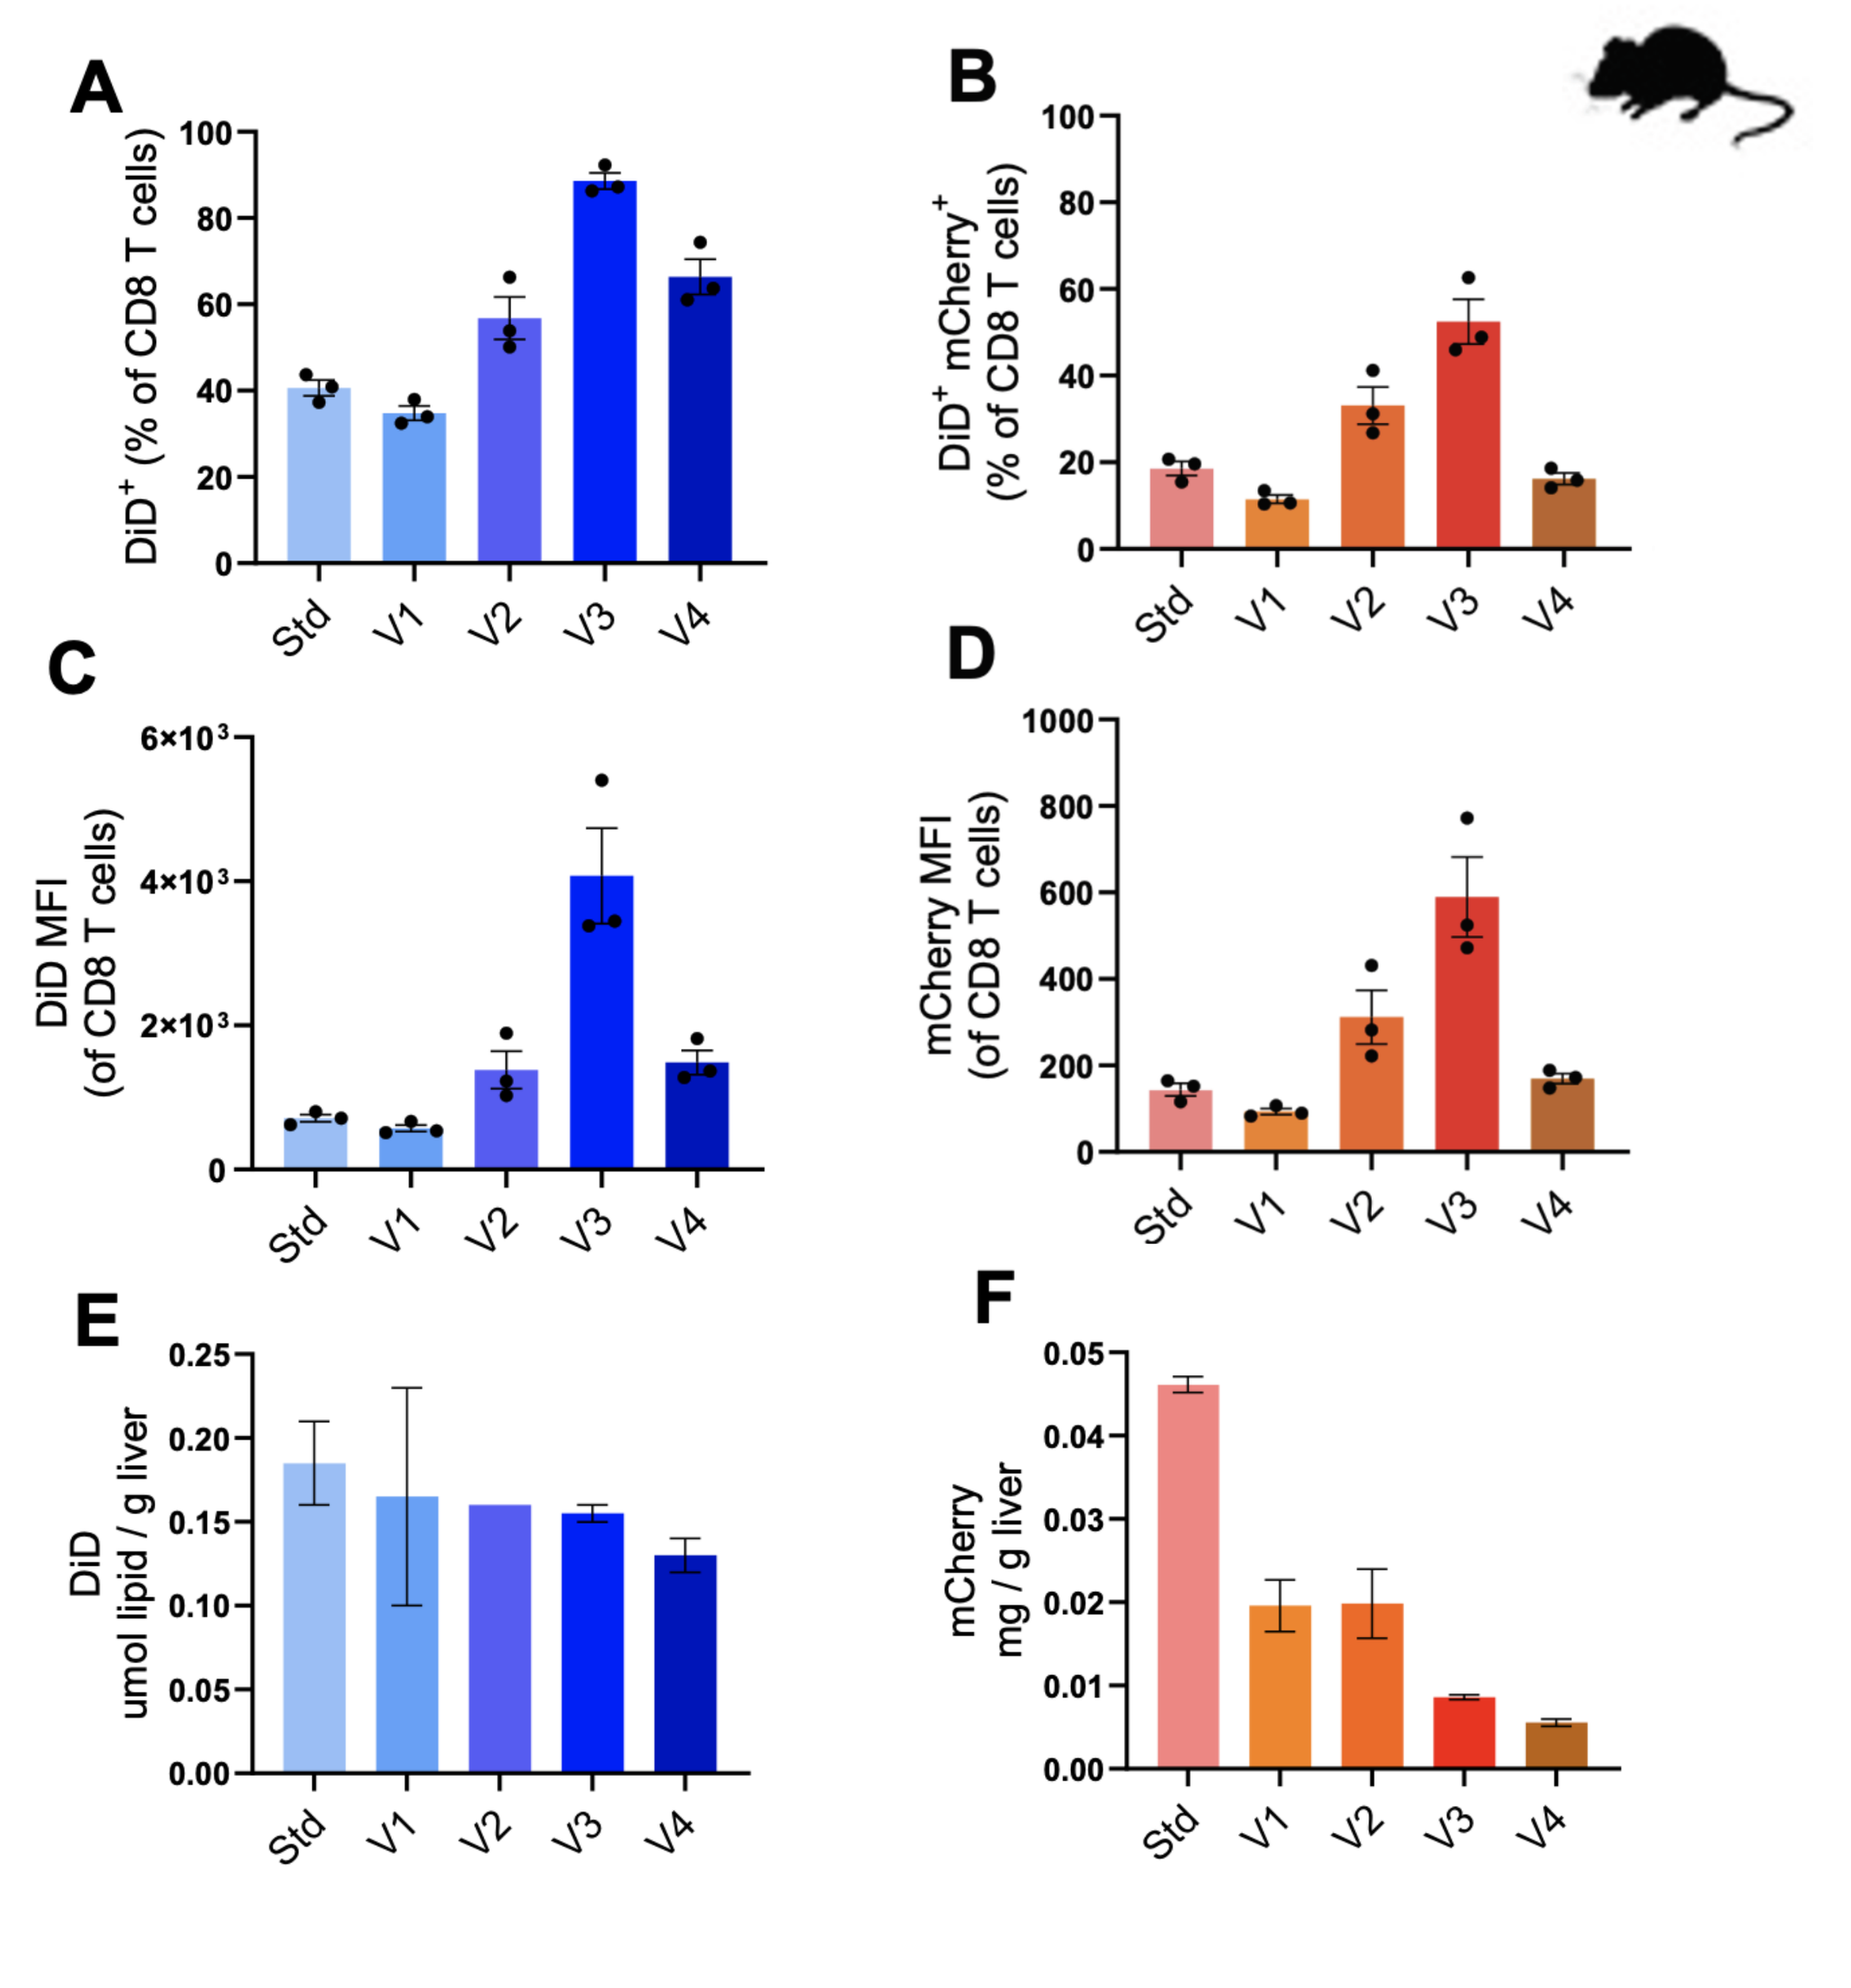
A
DiD⁺ (% of CD8 T cells)
Std V1 V2 V3 V4
B
DiD⁺ mCherry⁺ (% of CD8 T cells)
C
DiD MFI (of CD8 T cells)
D
mCherry MFI (of CD8 T cells)
E
DiD umol lipid / g liver
F
mCherry mg / g liver

Figure 5. Activity comparison of standard (Std) and long circulating variants.
% of LNP binding/uptake (**A**), RNA expression (**B**), MFI of binding (**C**) and expression (**D**) of splenic CD8 T cells after administration of DARPin-LNPs. LNP binding (**E**) and RNA expression (**F**) in liver tissue.

Storage stability of DARPin-LNP

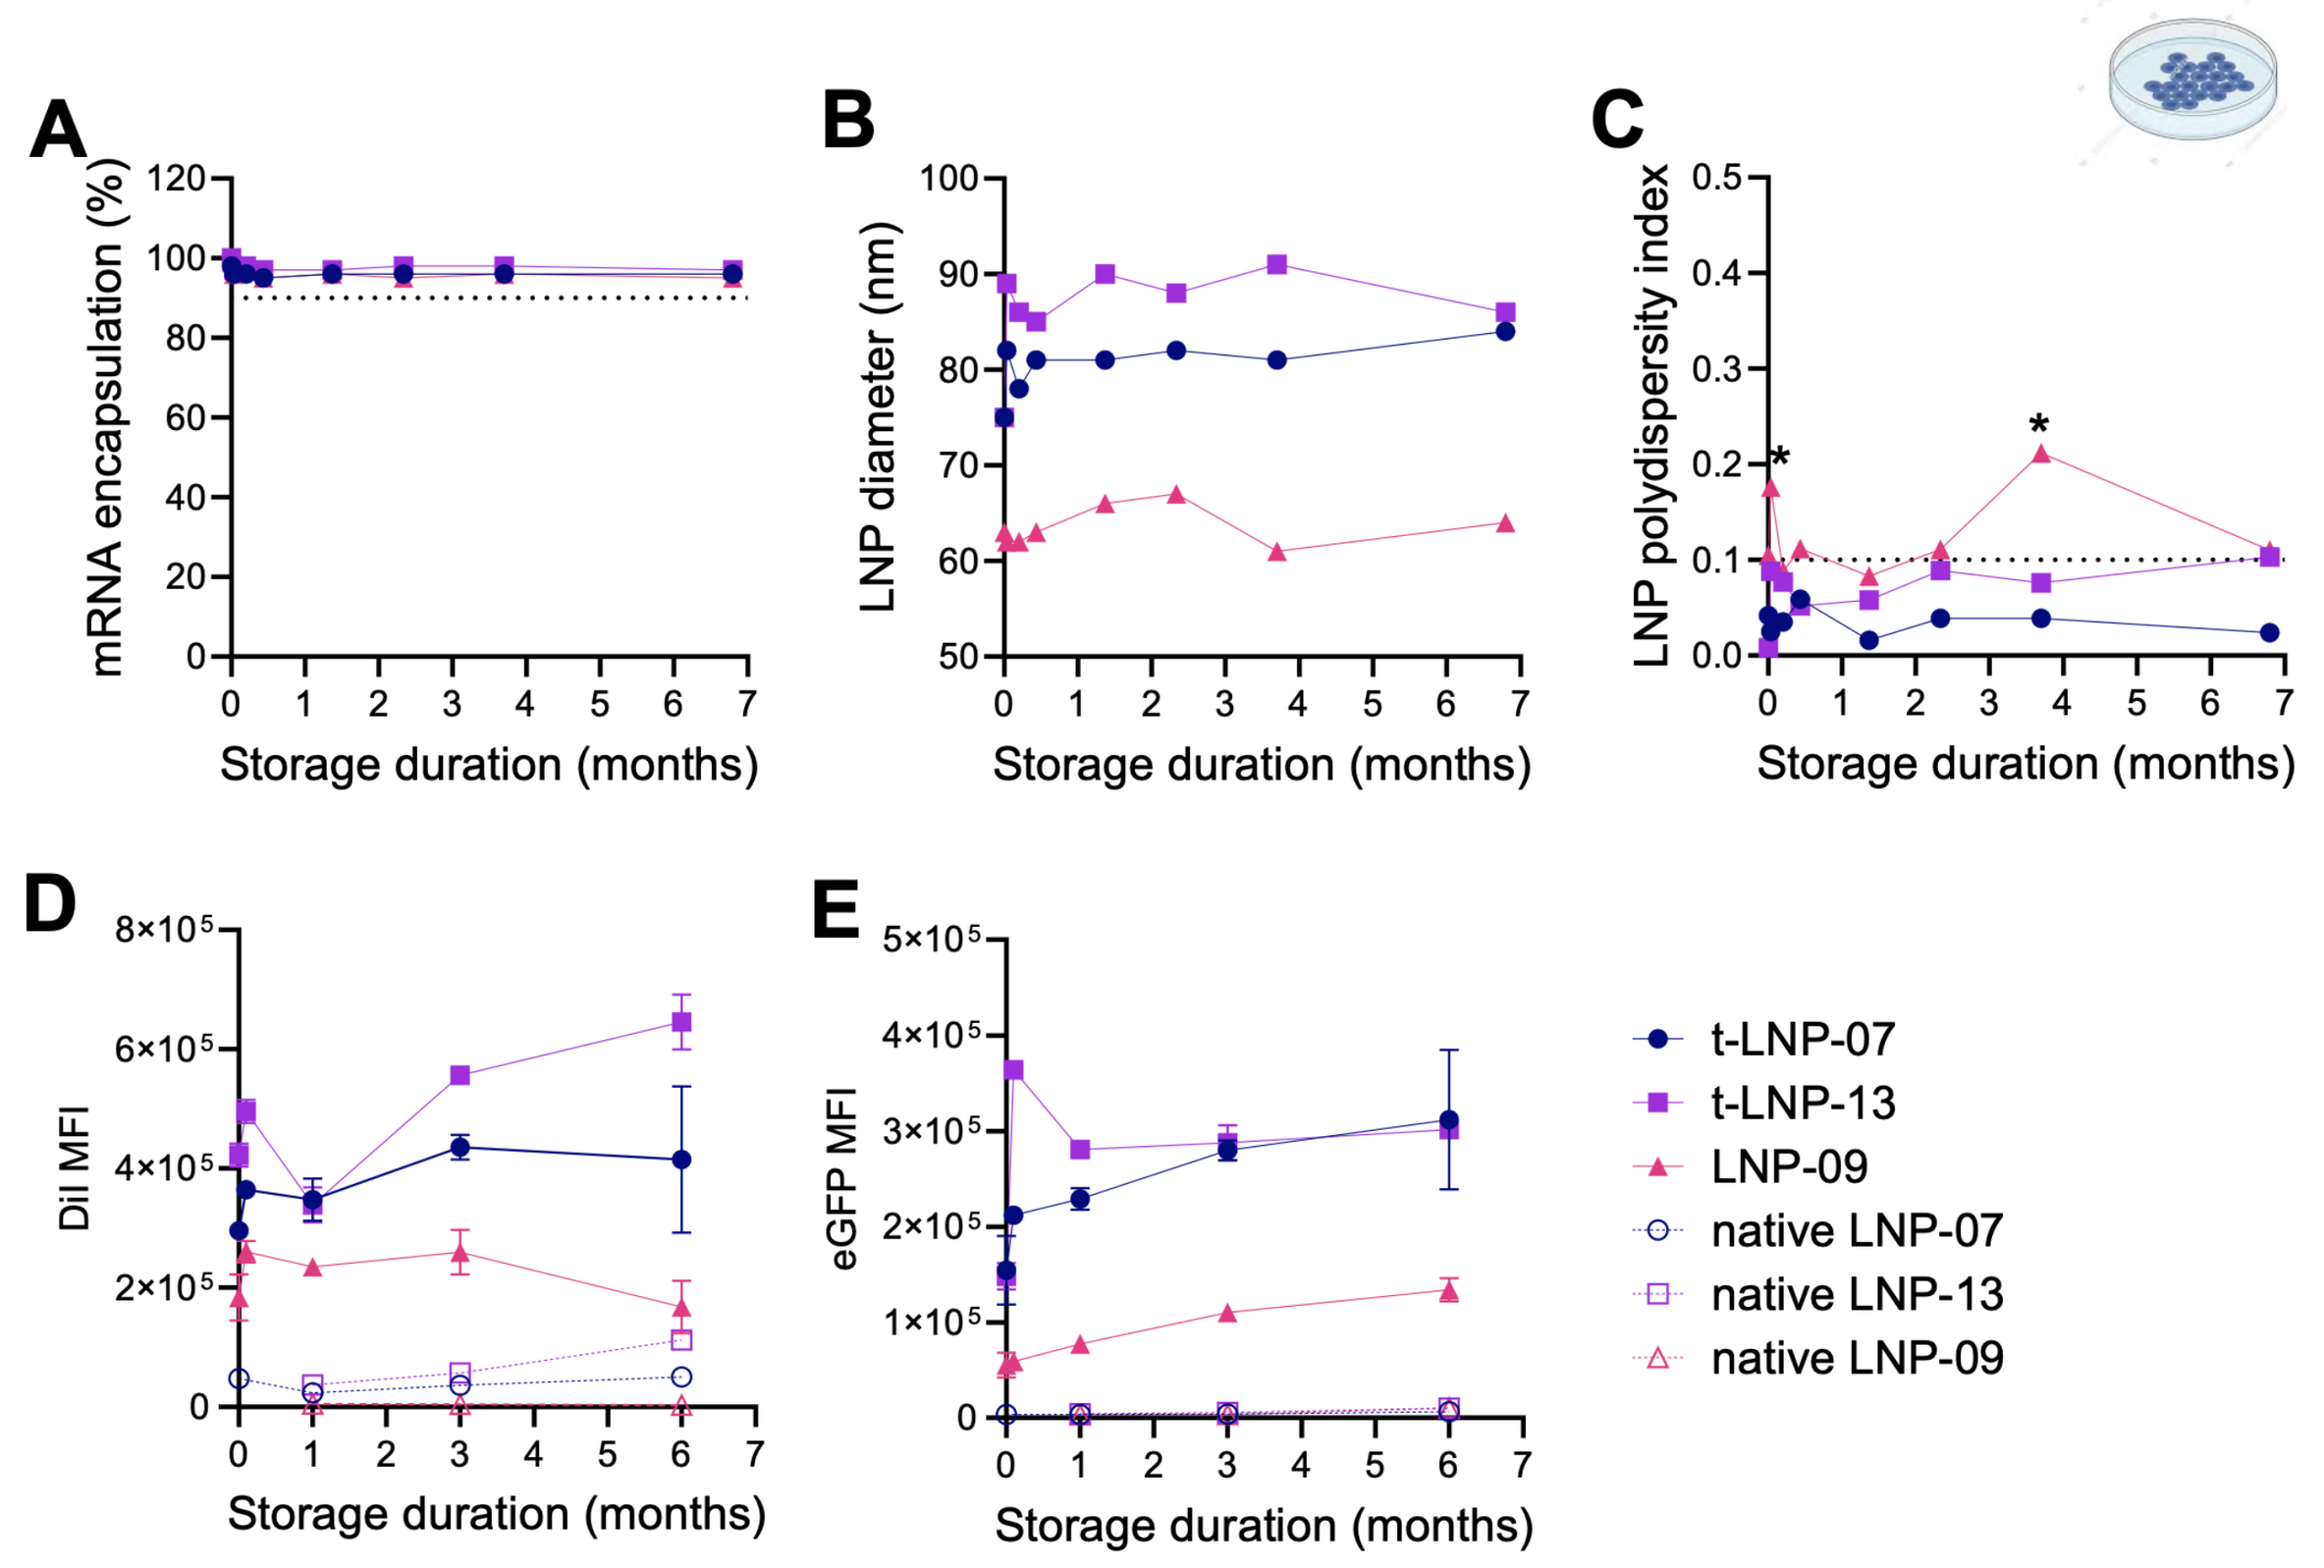
A
mRNA encapsulation (%)
Storage duration (months)
B
LNP diameter (nm)
C
LNP polydispersity index
D
DiI MFI
E
eGFP MFI
t-LNP-07
t-LNP-13
LNP-09
native LNP-07
native LNP-13
native LNP-09

Figure 6. Characteristics and activity of LNP after short and long storage.
RNA encapsulation % (**A**), LNP size (**B**), LNP polydispersity index (**C**), LNP binding/uptake (**D**) to and RNA expression (**E**) in SUP-T1 cell line up to 7 months storage at -80°C.

CD8 targeted binding/uptake and activity in human blood

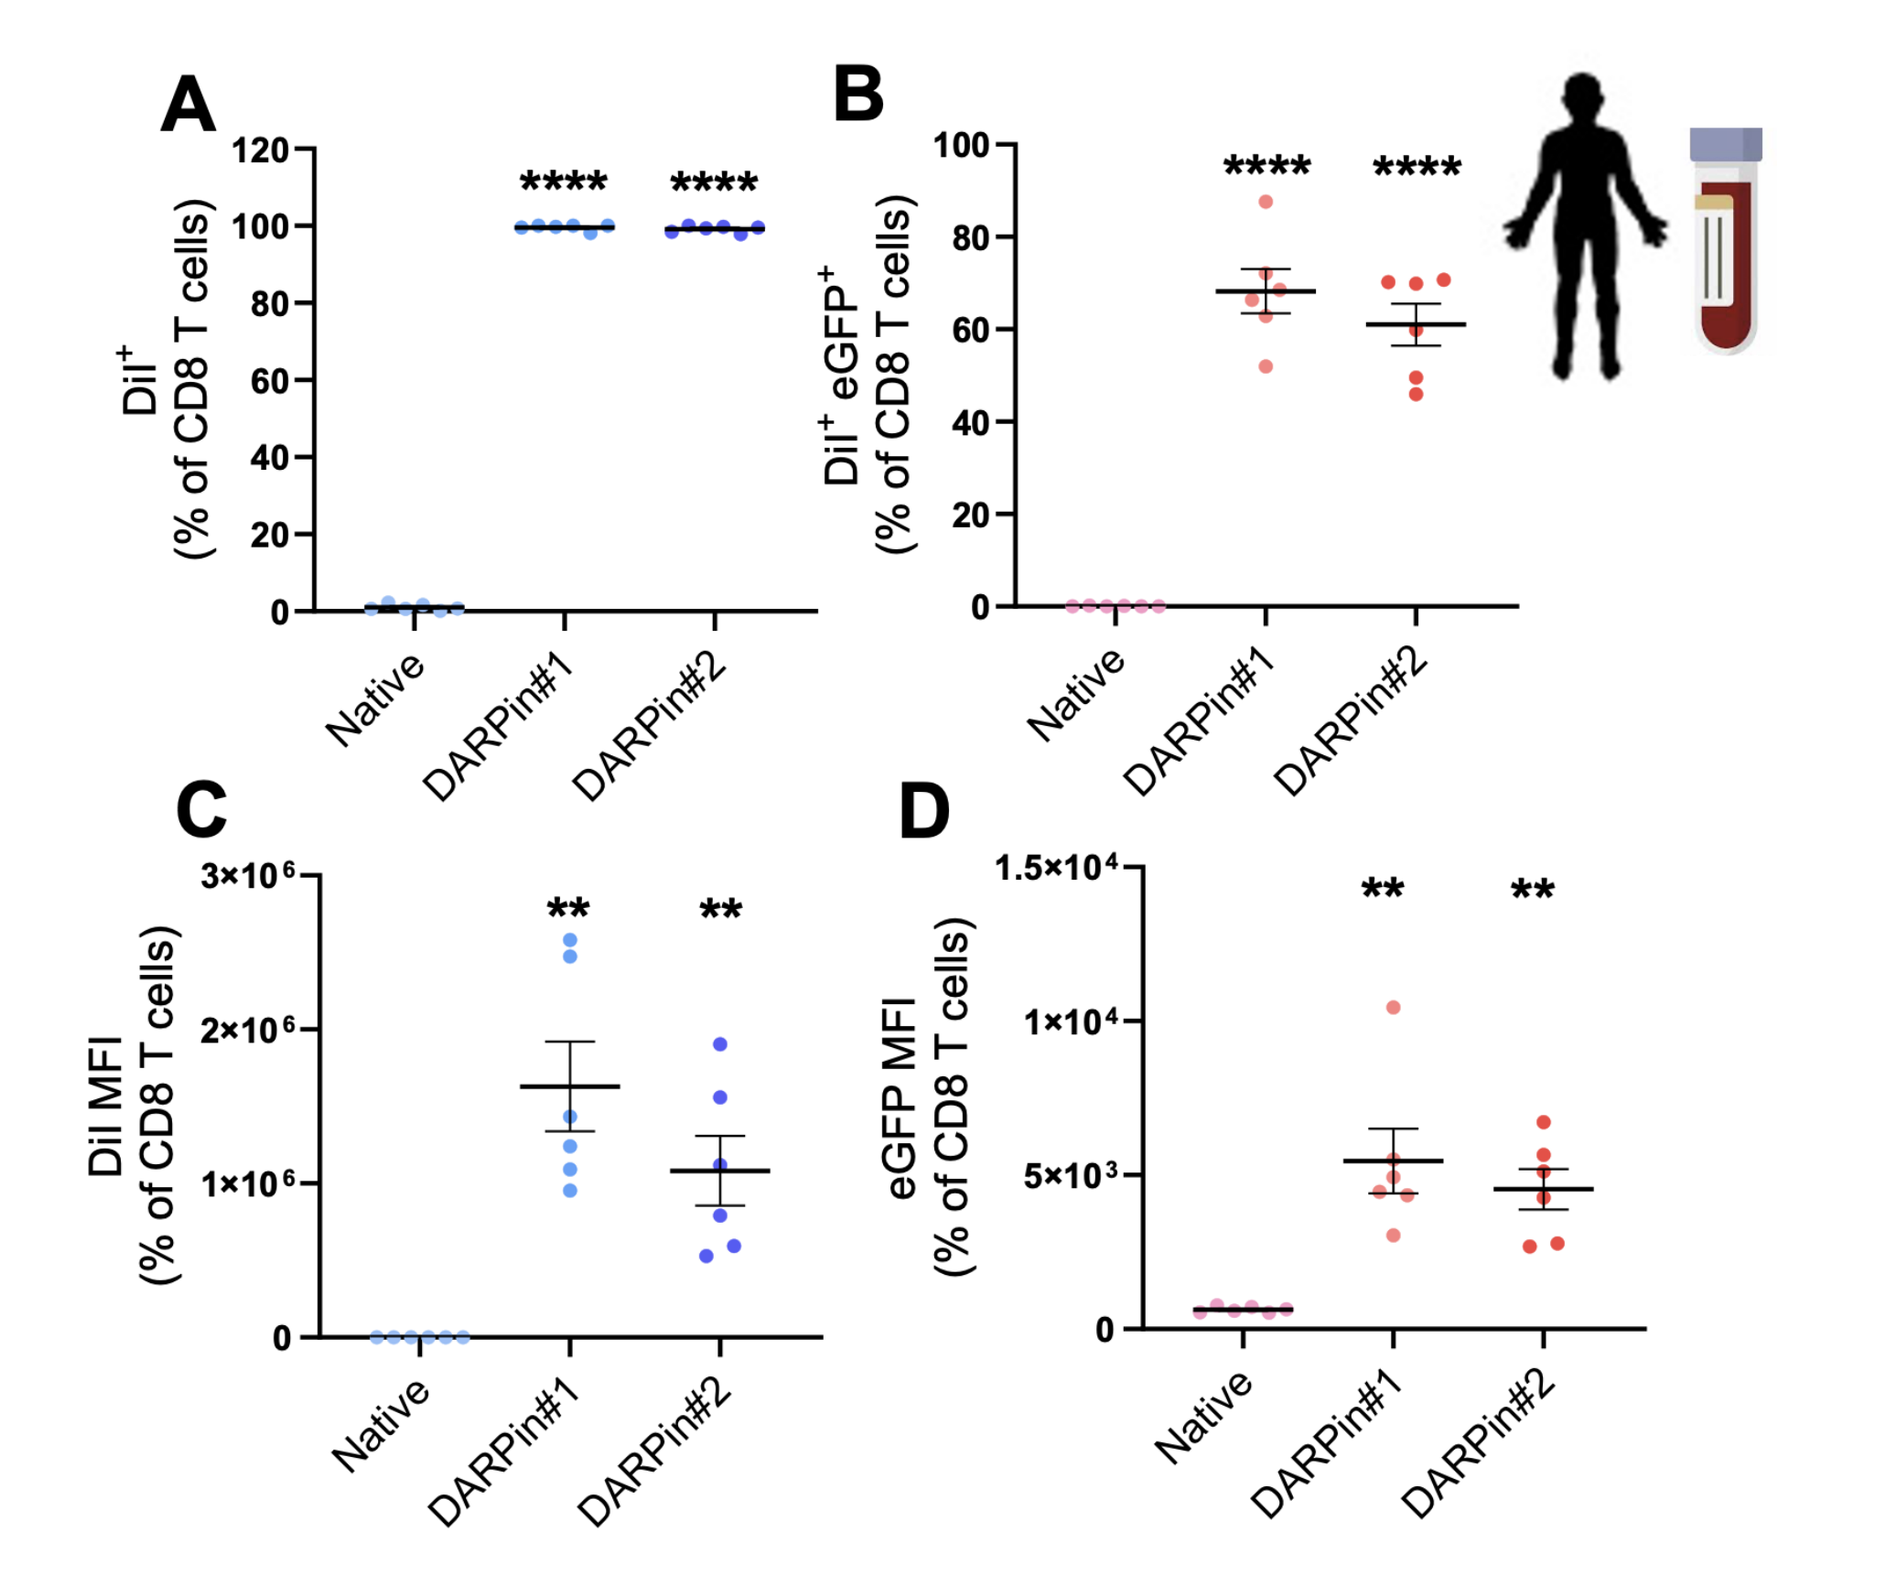
A
DiI⁺ (% of CD8 T cells)
Native DARPin#1 DARPin#2
B
DiI⁺ eGFP⁺ (% of CD8 T cells)
C
DiI MFI (% of CD8 T cells)
D
eGFP MFI (% of CD8 T cells)

Figure 7. DARPin-LNP application in human samples.
% of LNP binding/uptake (**A**), RNA expression (**B**), MFI of binding (**C**) to and expression (**D**) in human peripheral CD8 T cells after incubation ex vivo with human CD8 DARPin-LNPs.